Rapid Emergex Antigen Deployment Initiative (READI)

A synthetic T cell vaccine technology platform pairing a gold nanoparticle delivery system with select targets from a ligandome of prescreened pathogen derived, naturally processed & presented MHC-I peptides to provide rapid response solutions to combat and broadly protect against biothreat pathogens

EMERGEX VACCINES

GEORGE MASON UNIVERSITY | Biomedical Research Laboratory

Aykan Karabudak* | Xiaofang Huang | Brian Pfister
Emergex USA Corporation - Doylestown, PA
*Head of Immunology; ak@emergexvaccines.com

Aarthi Narayanan | Farhang Alem
Biomedical Research Laboratory
George Mason University - Fairfax, VA

READI Platform Core Technologies

The **READI** platform is a **fully synthetic** pairing of **gold nanoparticles** with **naturally processed & presented** pathogen-specific **peptide** antigens to contribute to a broadly **protective cellular immune response**.

Microneedle delivery enhances the **MHC Class I** immune subsystem eliciting **antigen-specific, functional T cells** in addition to **long-term protection**.

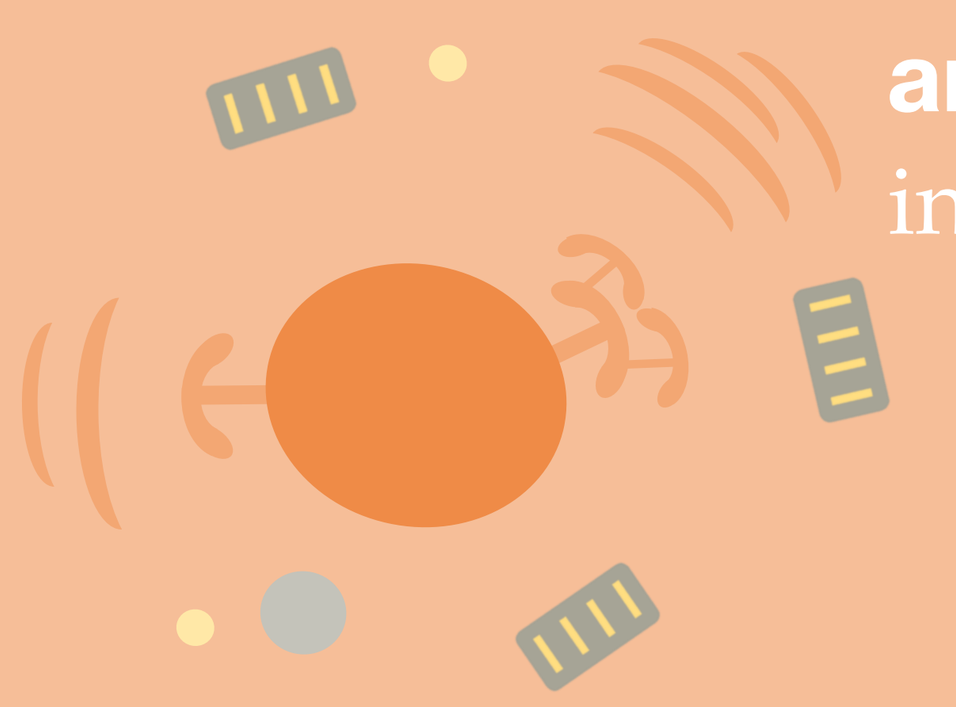

Mass Spectrometry Ligandome Discovery

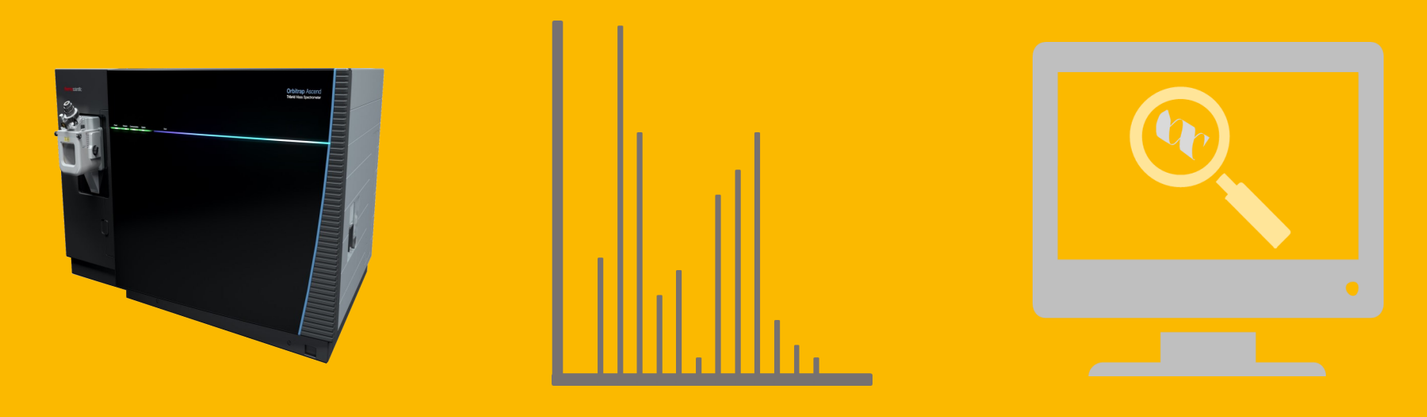

Fully Synthetic Gold Nanoparticle Vehicle

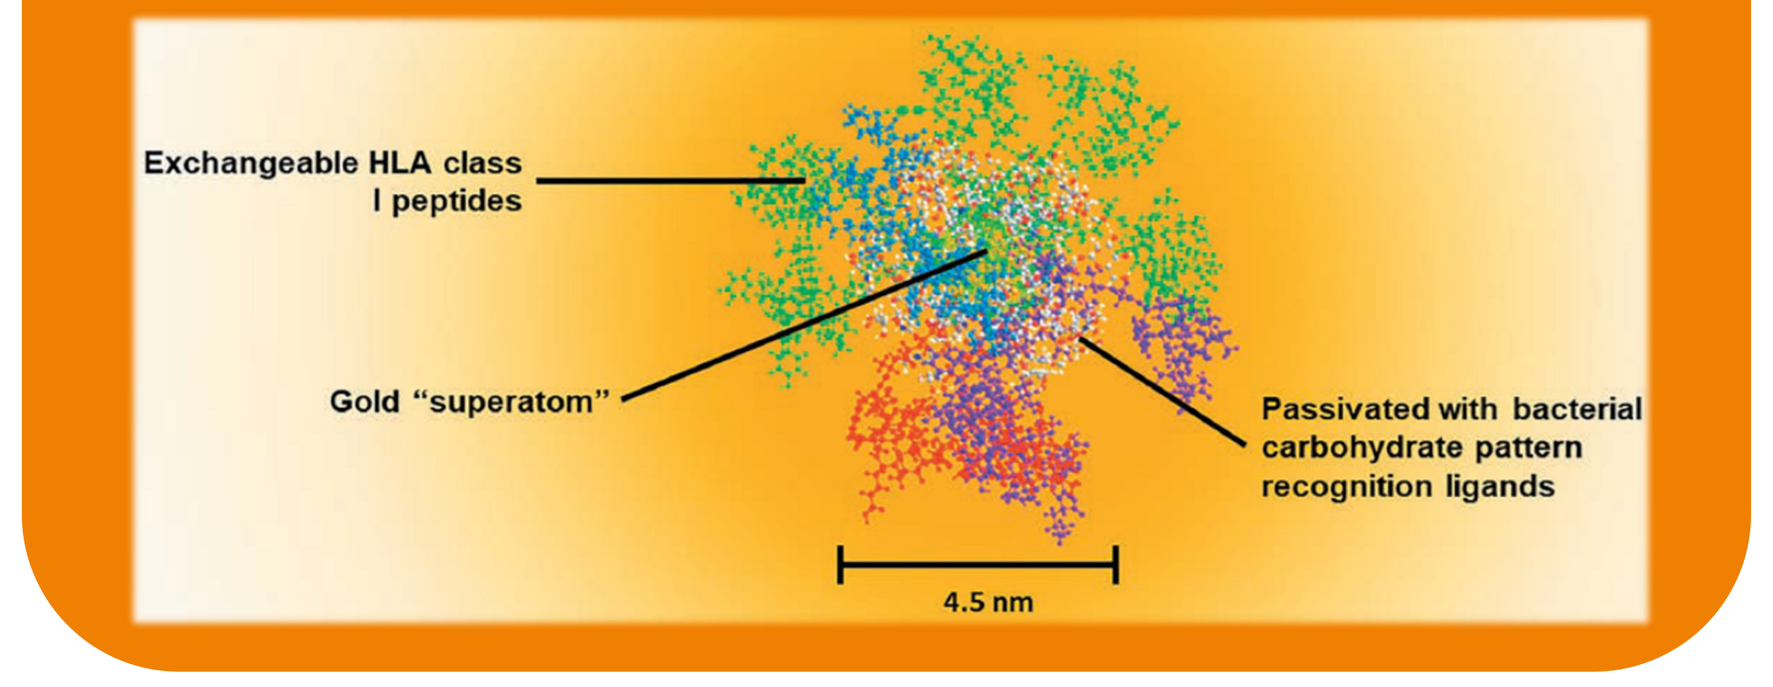

Exchangeable HLA class I peptides
Gold "superatom"
Passivated with bacterial carbohydrate pattern recognition ligands
4.5 nm

Emergex's **T cell adaptive vaccines** evoke the course of cellular immunity typically prompted in **natural infection**

Microneedle Array Patch Technology

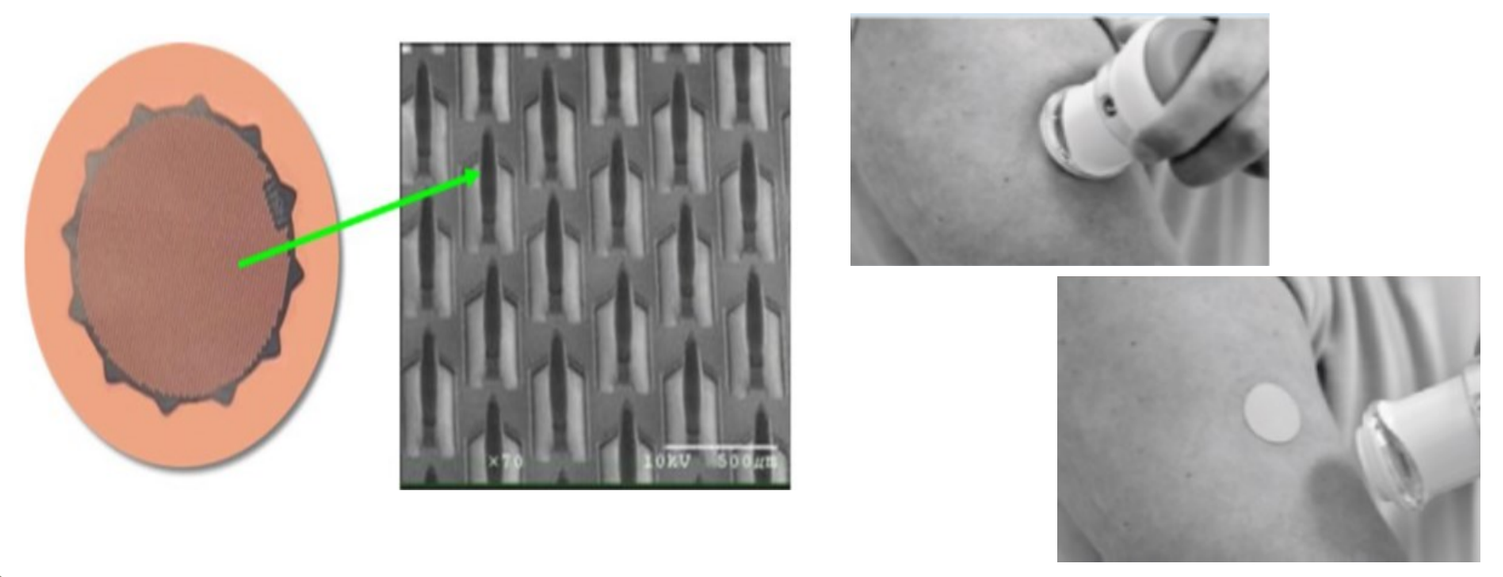

READI Platform Vaccine Formulations

Targeting a Diversity of Pathogens

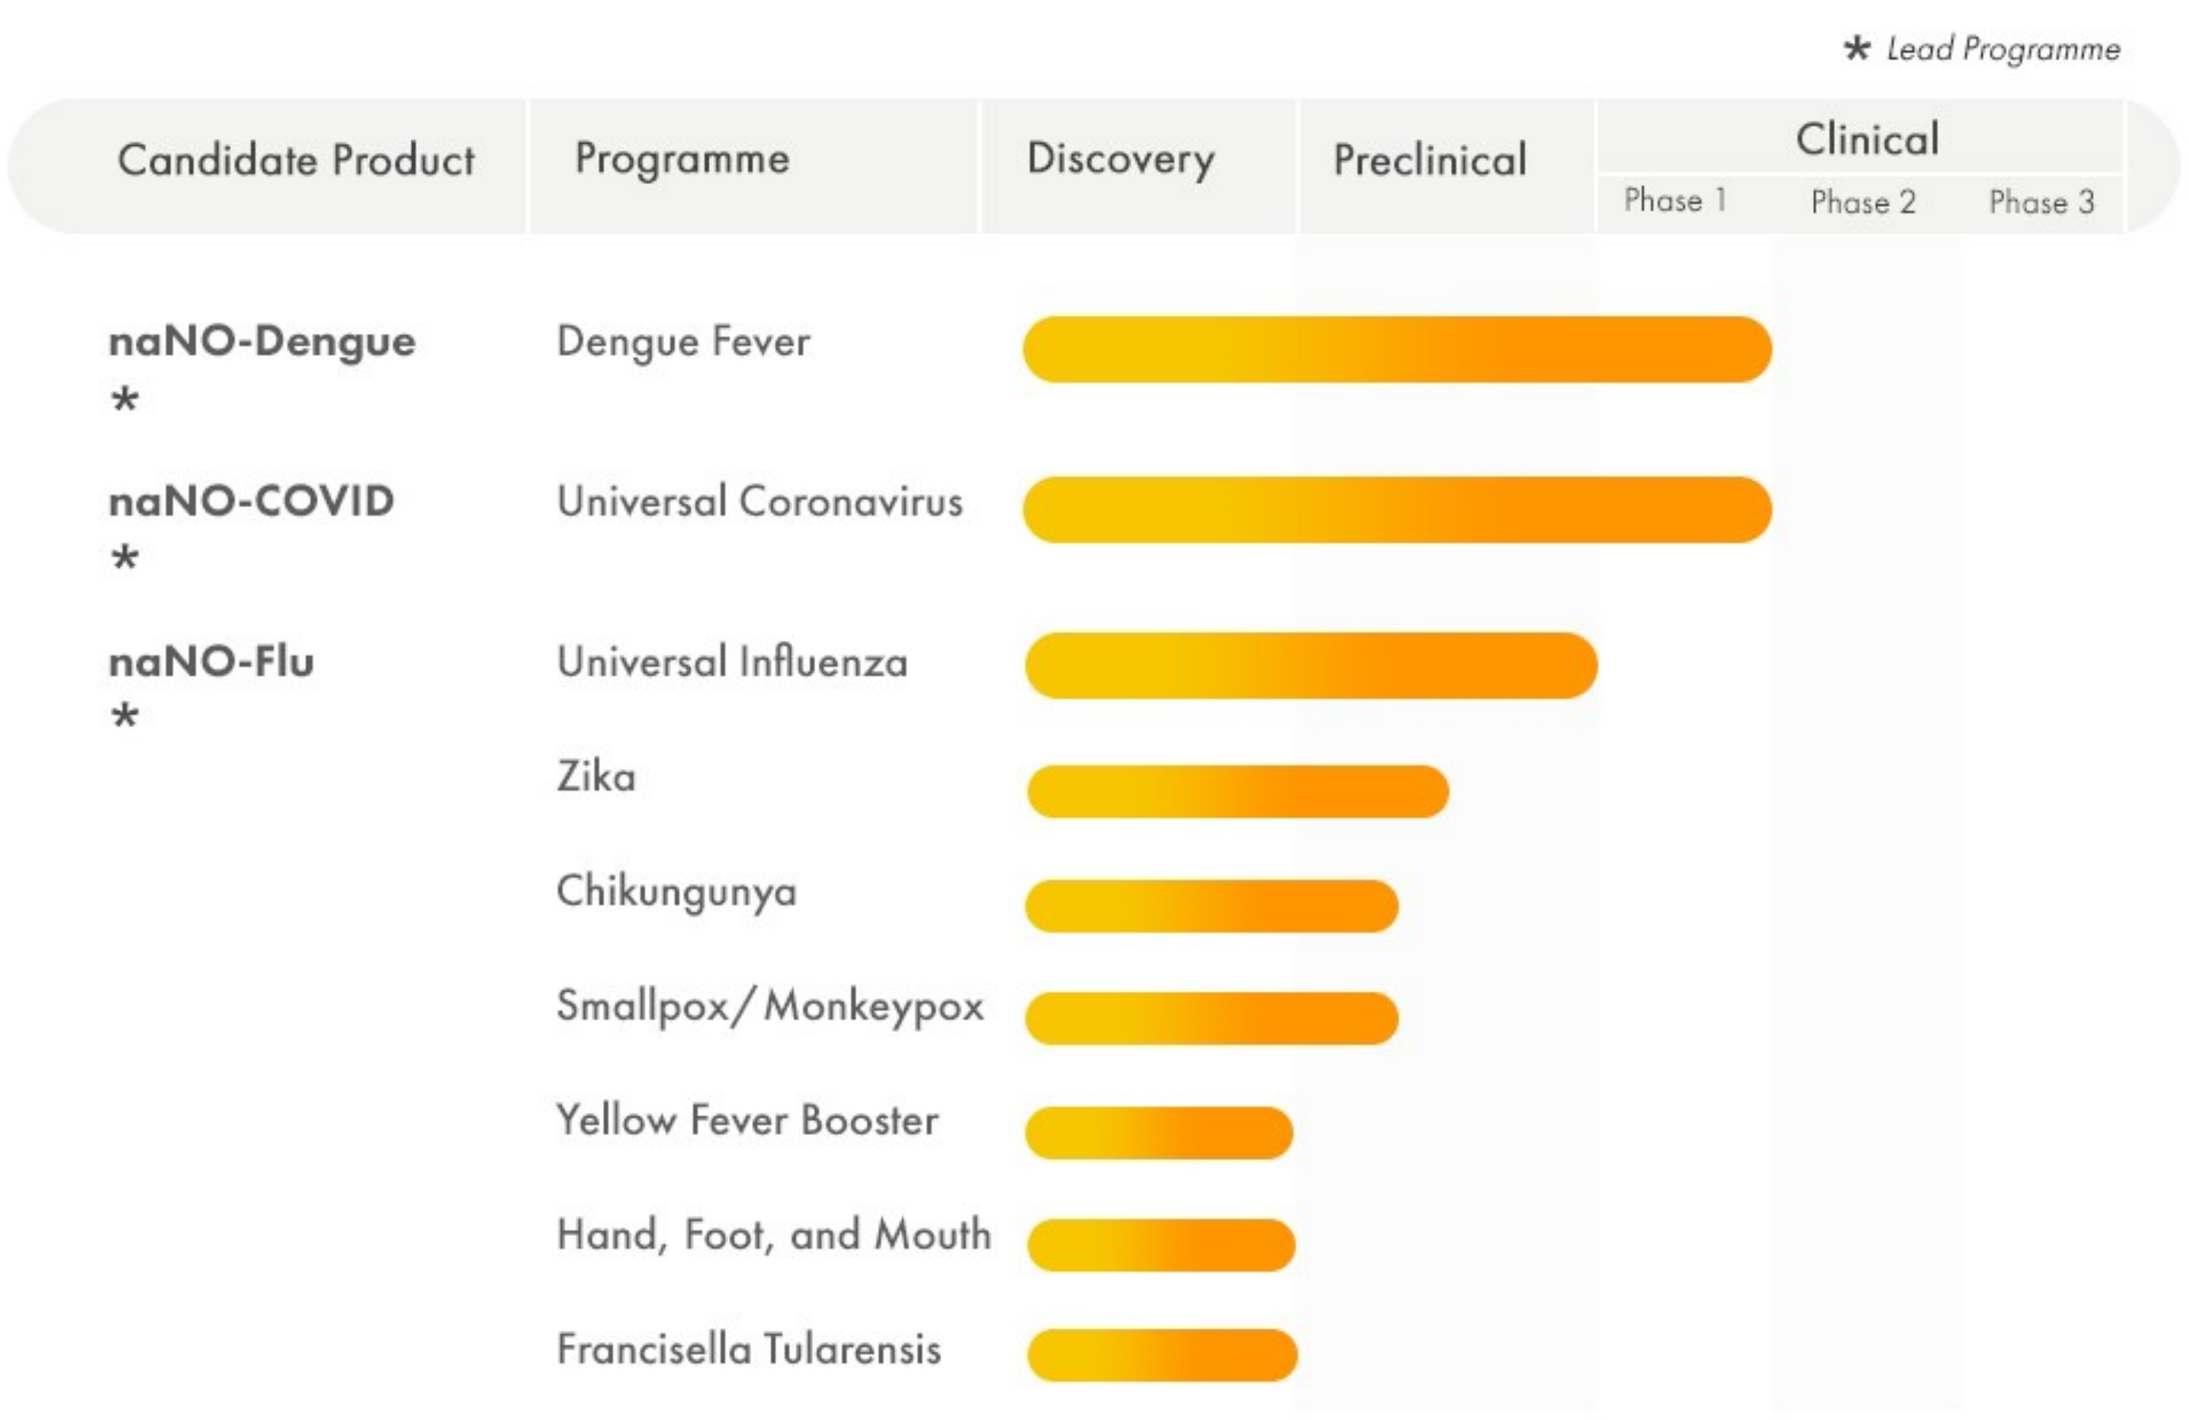

★ Lead Programme
Candidate Product
Programme
Discovery
Preclinical
Clinical
Phase 1
Phase 2
Phase 3
naNO-Dengue *
Dengue Fever
naNO-COVID *
Universal Coronavirus
naNO-Flu *
Universal Influenza
Zika
Chikungunya
Smallpox/ Monkeypox
Yellow Fever Booster
Hand, Foot, and Mouth
Francisella Tularensis

Prescreened **MHC Class I Pathogen-specific Ligandomes** can be positioned for **Biothreat Preparedness**

Simple & **rapid** manufacture yields **100% synthetic**, shelf-stable **vaccine formulations**

Targeted microneedle delivery of **T cell Adaptive Vaccine** elicits **broad, long-term CD8+ T cell protection** against pathogens

READI Platform Technology Pre-clinical Foundations

Naturally processed & presented infection model paired with novel immunoproteomic technique provides for the empirical determination of MHC Class-I T cell pathogen-specific epitope candidates

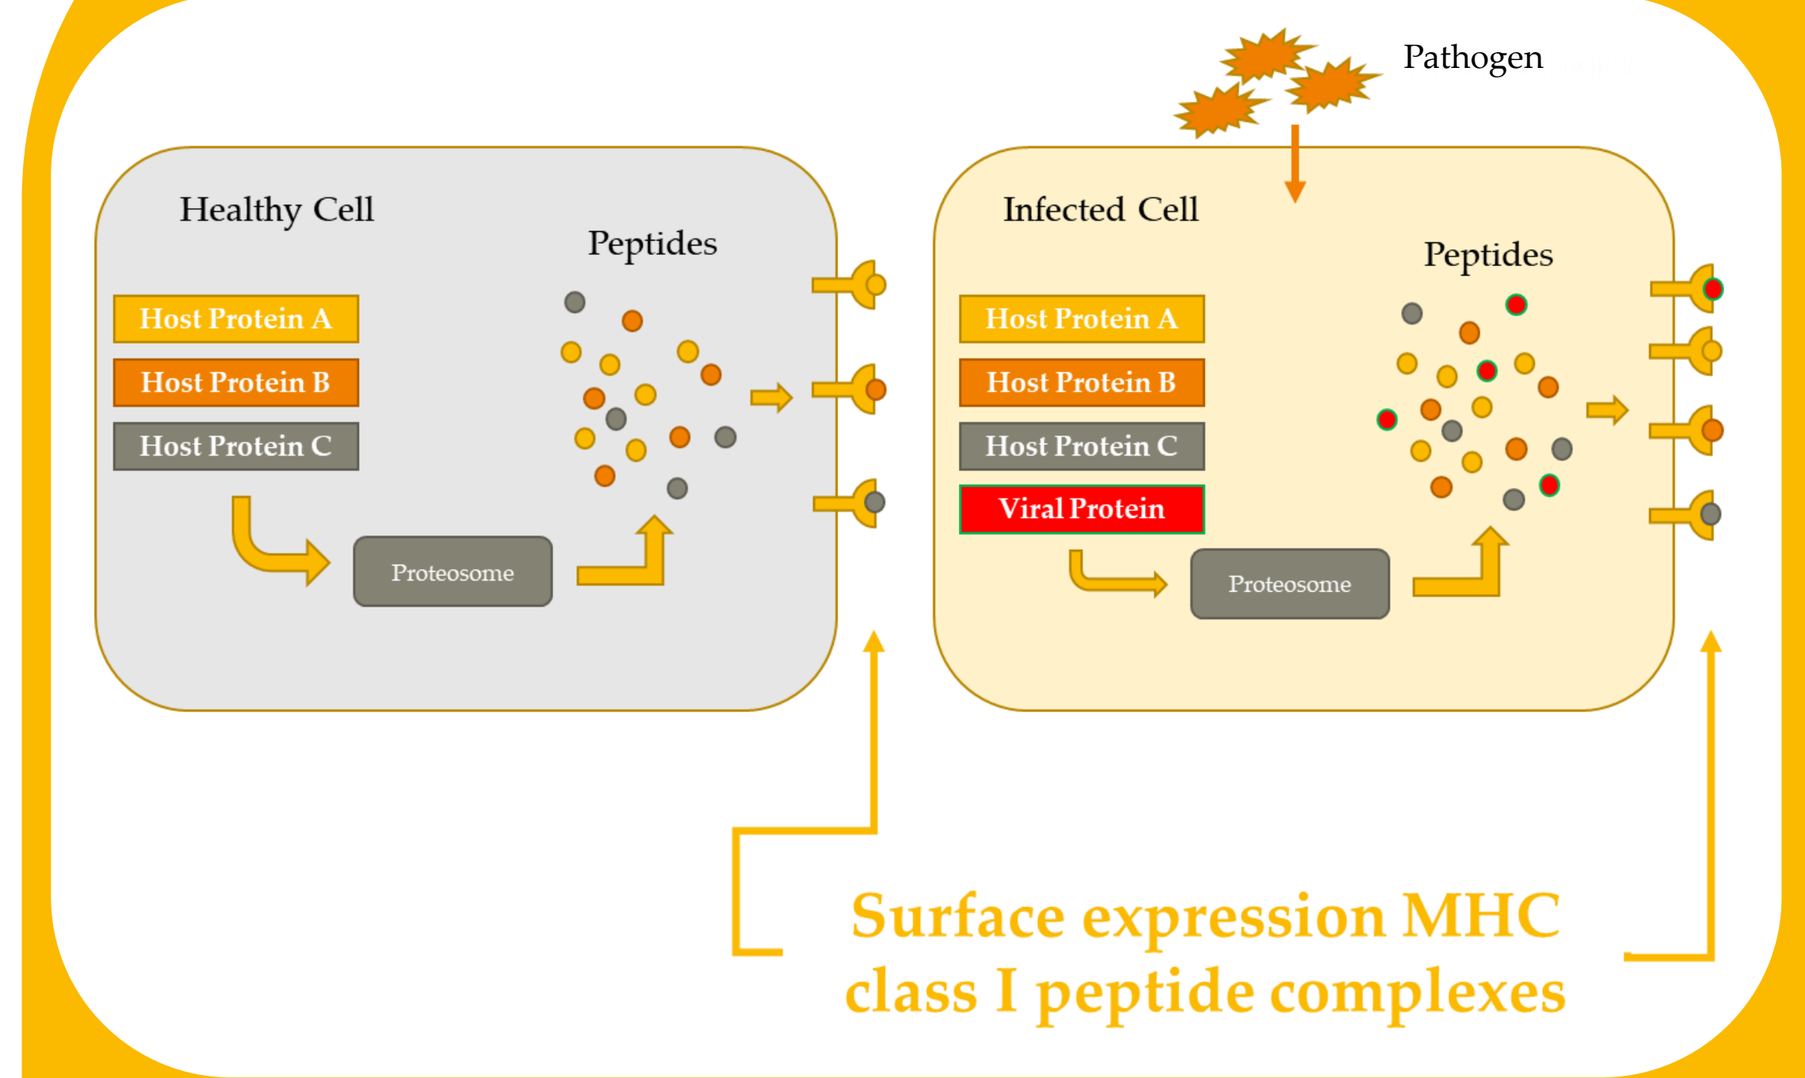

Pathogen
Healthy Cell
Peptides
Host Protein A
Host Protein B
Host Protein C
Proteosome
Infected Cell
Peptides
Host Protein A
Host Protein B
Host Protein C
Viral Protein
Proteosome
Surface expression MHC class I peptide complexes

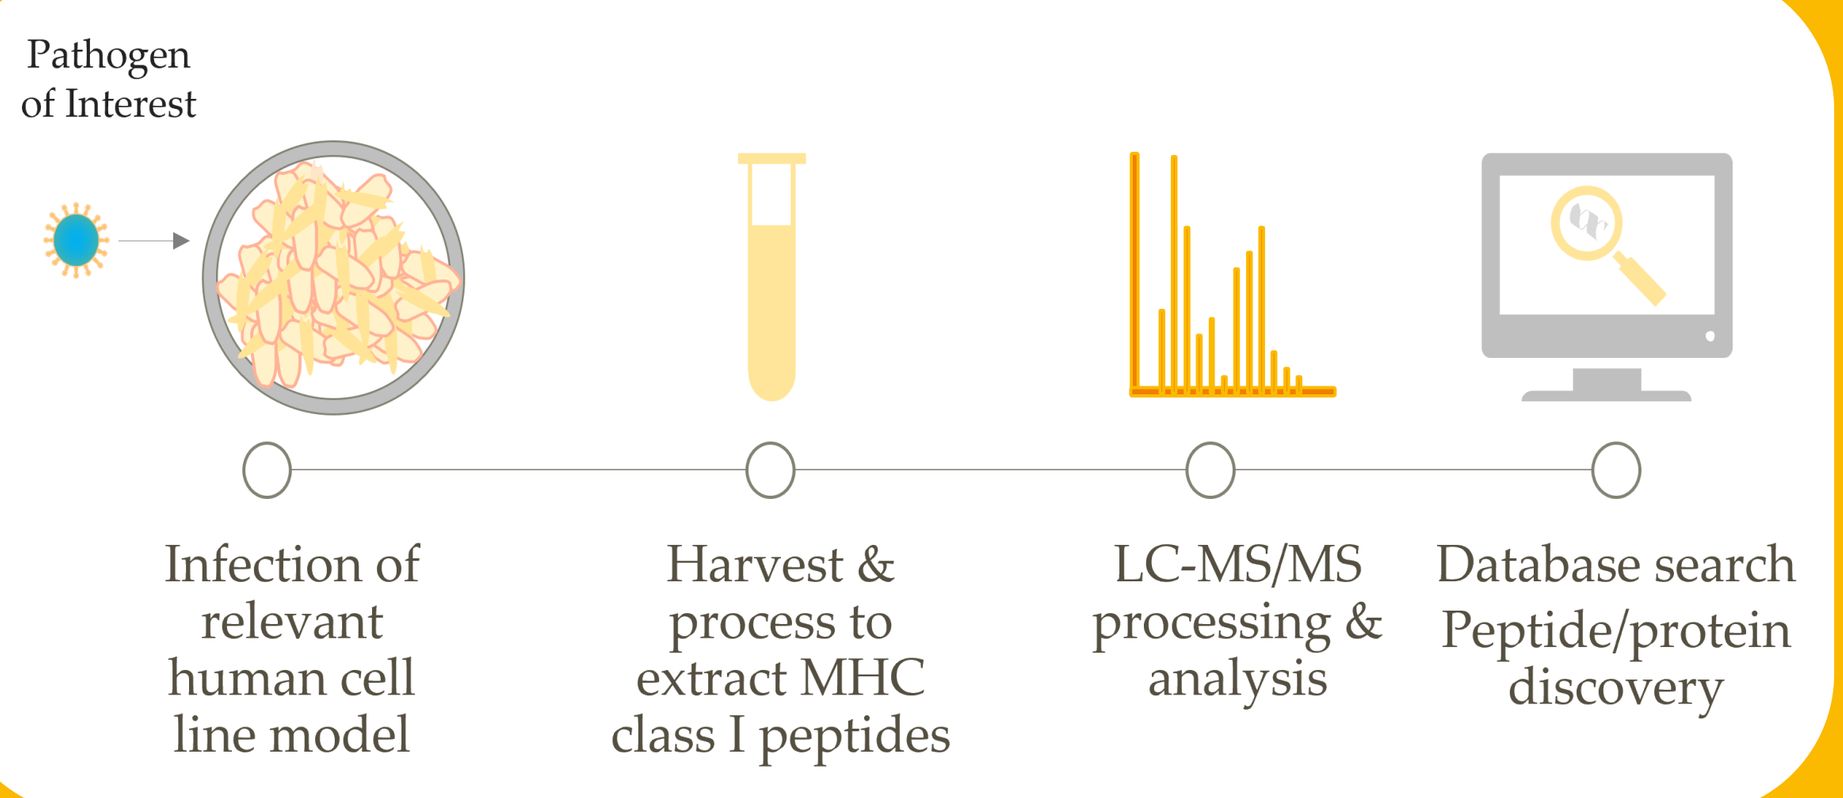

Pathogen of Interest
Infection of relevant human cell line model
Harvest & process to extract MHC class I peptides
LC-MS/MS processing & analysis
Database search Peptide/protein discovery

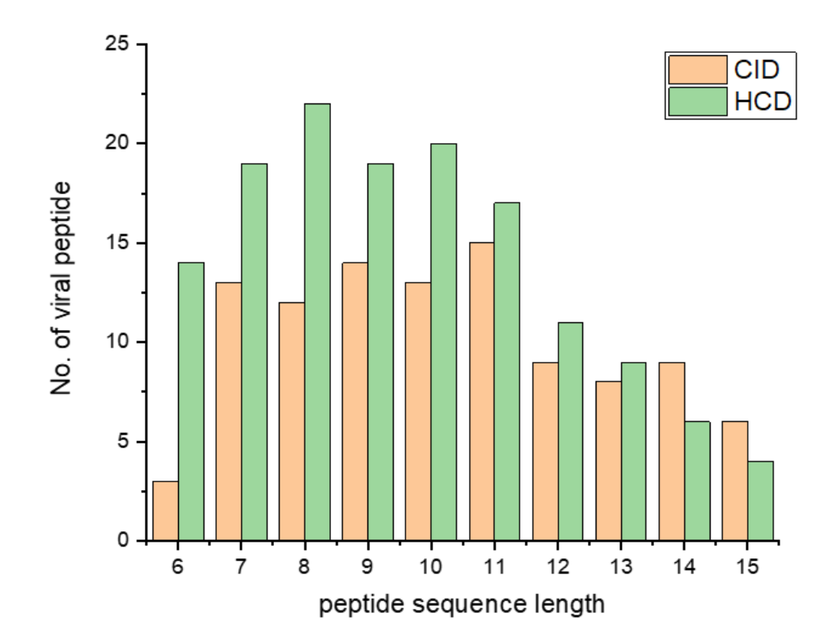

CID
HCD
No. of viral peptide
peptide sequence length

Distribution of CHIKV peptides via different fragmentation methods

Criterion Analysis for Candidate Selection

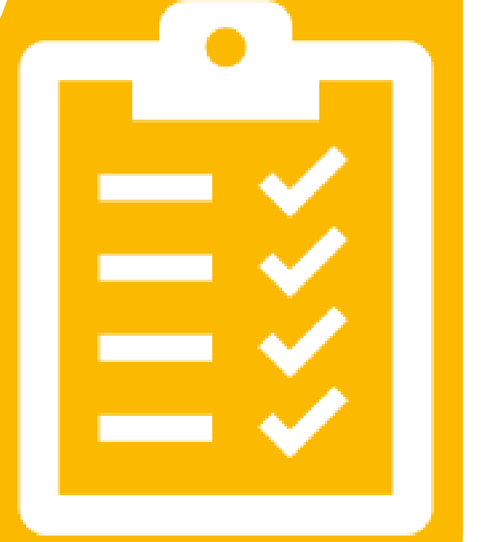

The Prescreened Ligandome

Generation of multivalent & multiallelic T Cell Adaptive vaccine formulations utilizing the pathogen-specific Ligandome candidate epitopes paired with gold nanoparticle (GNP) core

Emergex vaccines are multivalent and multiallelic providing broad genetic population coverage while ensuring longevity of efficacy

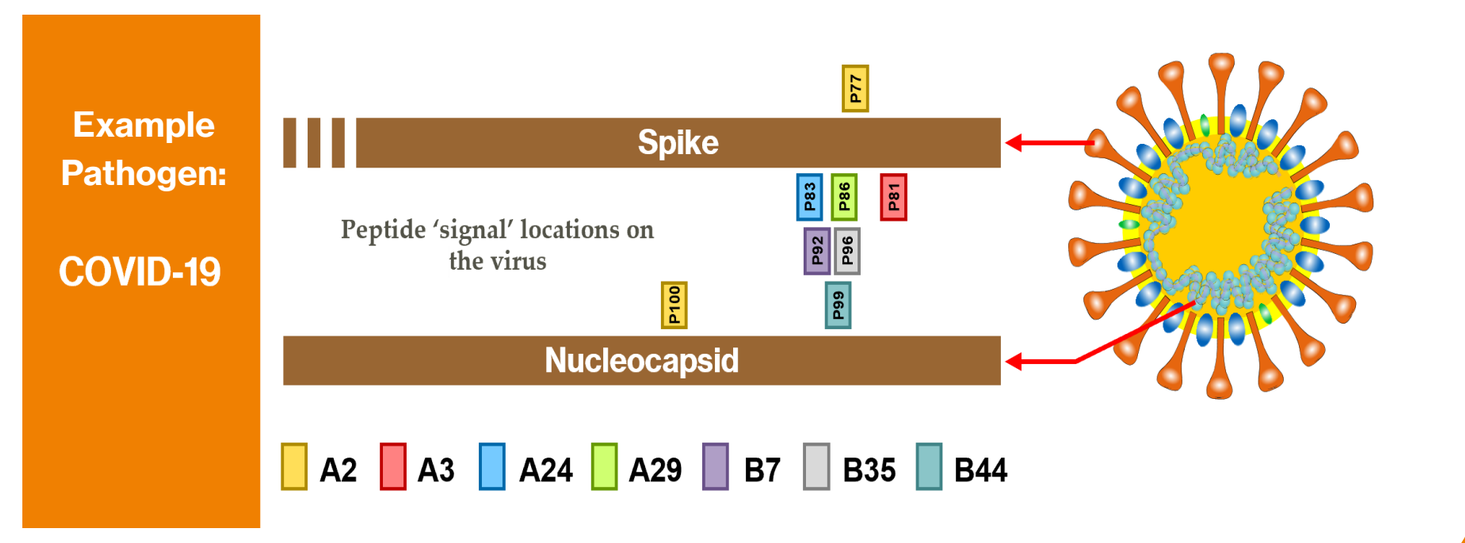

Example Pathogen:
COVID-19
Spike
Peptide 'signal' locations on the virus
Nucleocapsid
P77
P83
P86
P81
P92
P96
P99
P100
A2
A3
A24
A29
B7
B35
B44

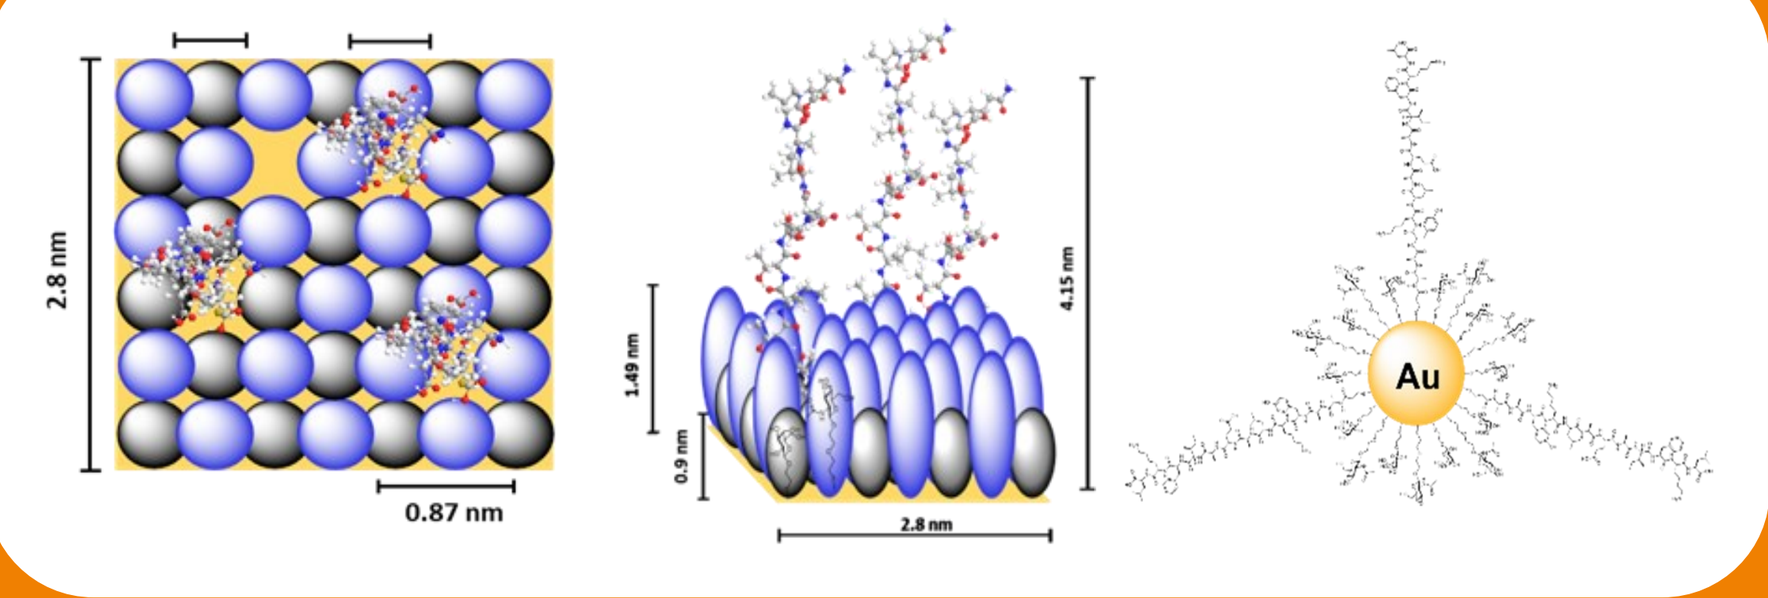

2.8 nm
0.87 nm
1.49 nm
0.9 nm
2.8 nm
4.15 nm
Au

- Gold core particle overcomes T cell vaccine hurdles including **inhibition of enzymatic activity** at surface layer of the superatom & provides payload stabilization
- **Fully Synthetic**, ease of manufacture, targeted delivery of intradermal layers
- **Adjuvant characteristics** due to bacterial mimetic carbohydrates as known pattern recognition elements, which also render the particle **water soluble**
- By using **'conserved' peptides** from various proteins as well as conserved regions of the virus, **the impact of viral mutations is reduced**
- Multiallelic **HLA class I specific peptides** are included in Emergex's vaccine candidates — covering multiple HLA alleles helps broad population coverage

Delivery & Proper Processing, Presentation of Bio-Payload using GNP

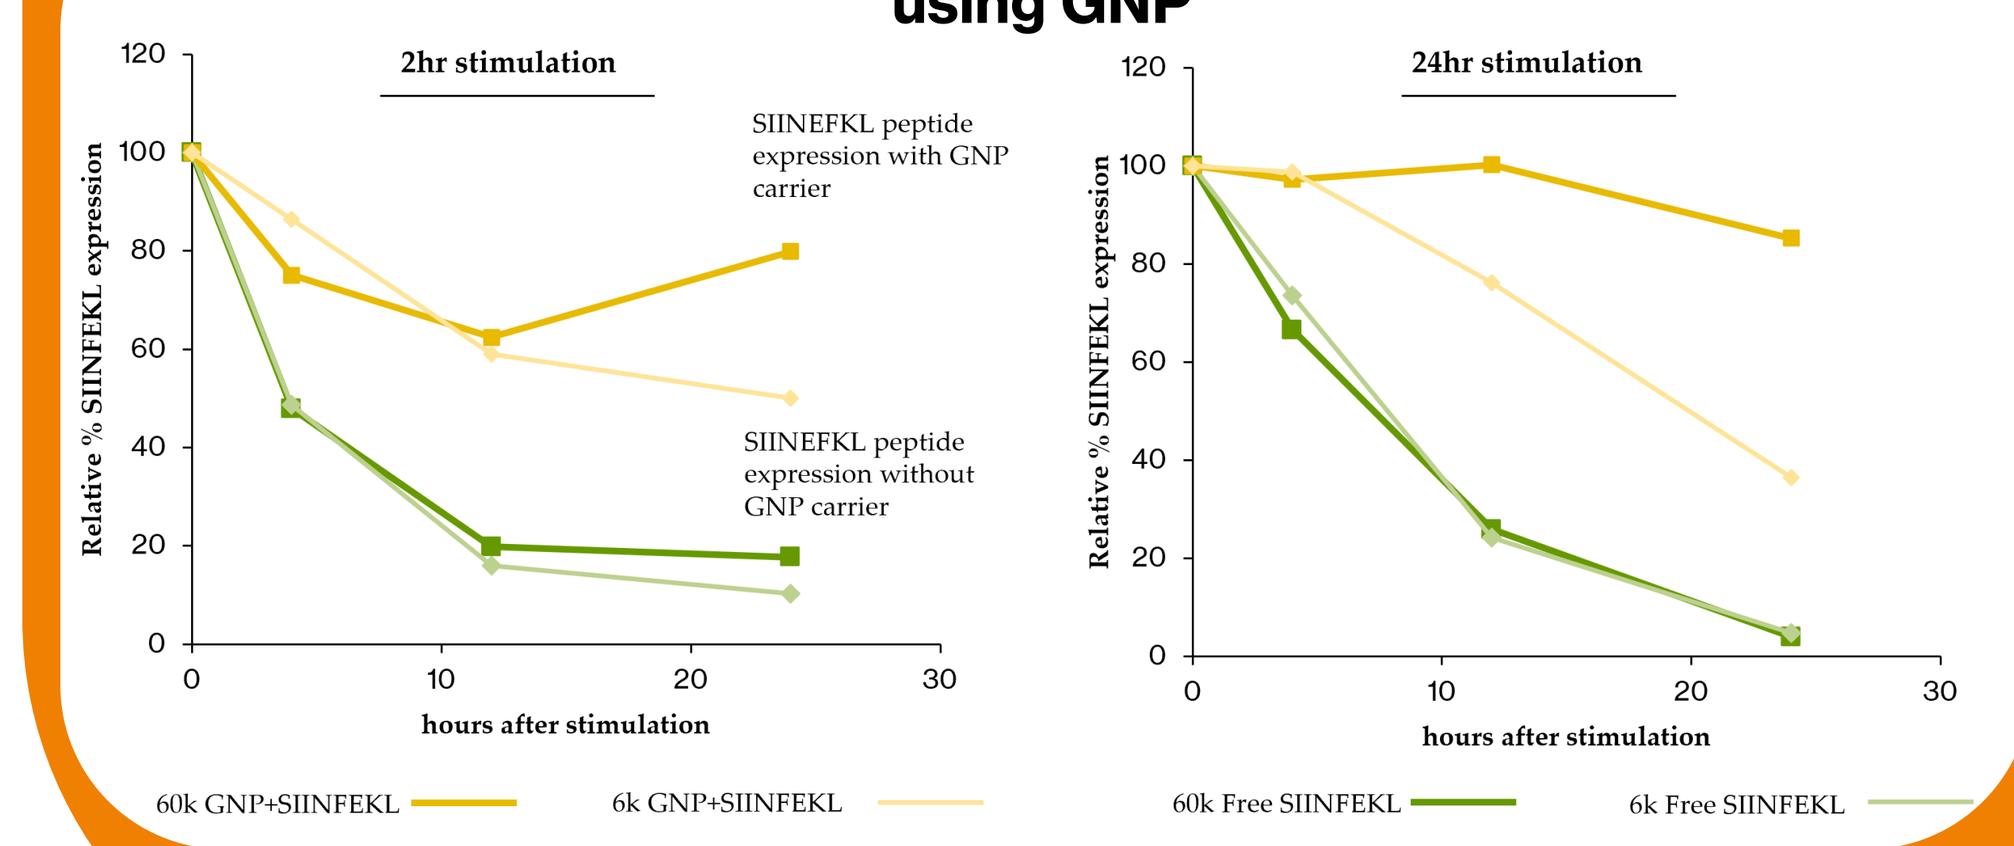

2hr stimulation
SIINEFKL peptide expression with GNP carrier
SIINEFKL peptide expression without GNP carrier
Relative % SIINFEKL expression
hours after stimulation
24hr stimulation
Relative % SIINFEKL expression
hours after stimulation
60k GNP+SIINFEKL
6k GNP+SIINFEKL
60k Free SIINFEKL
6k Free SIINFEKL

Delivering the Adaptive T Cell Vaccine formulation via microneedles targets specialized immune cells to elicit functional, long-term CD8+ T cell protection

- **Emergex's vaccine** formulations mimic **natural infections**, delivering synthetic MHC class I viral peptides to APCs which 'prime' **CD8+ T cells**
- Initial vaccination leads to development of circulating **T_{EM} cells** with tissue-homing potential.
- **Secondary vaccination** seeds viral class I peptides in the tissues to achieve antigen recognition **sans-natural infection** including signalling distal tissue DCs to present antigens to circulating **T_{EM} cells** signalling **differentiation** to **T_{RM} cells**, which are able to abort an infection prior to disease pathogenesis & have been shown to be active for decades

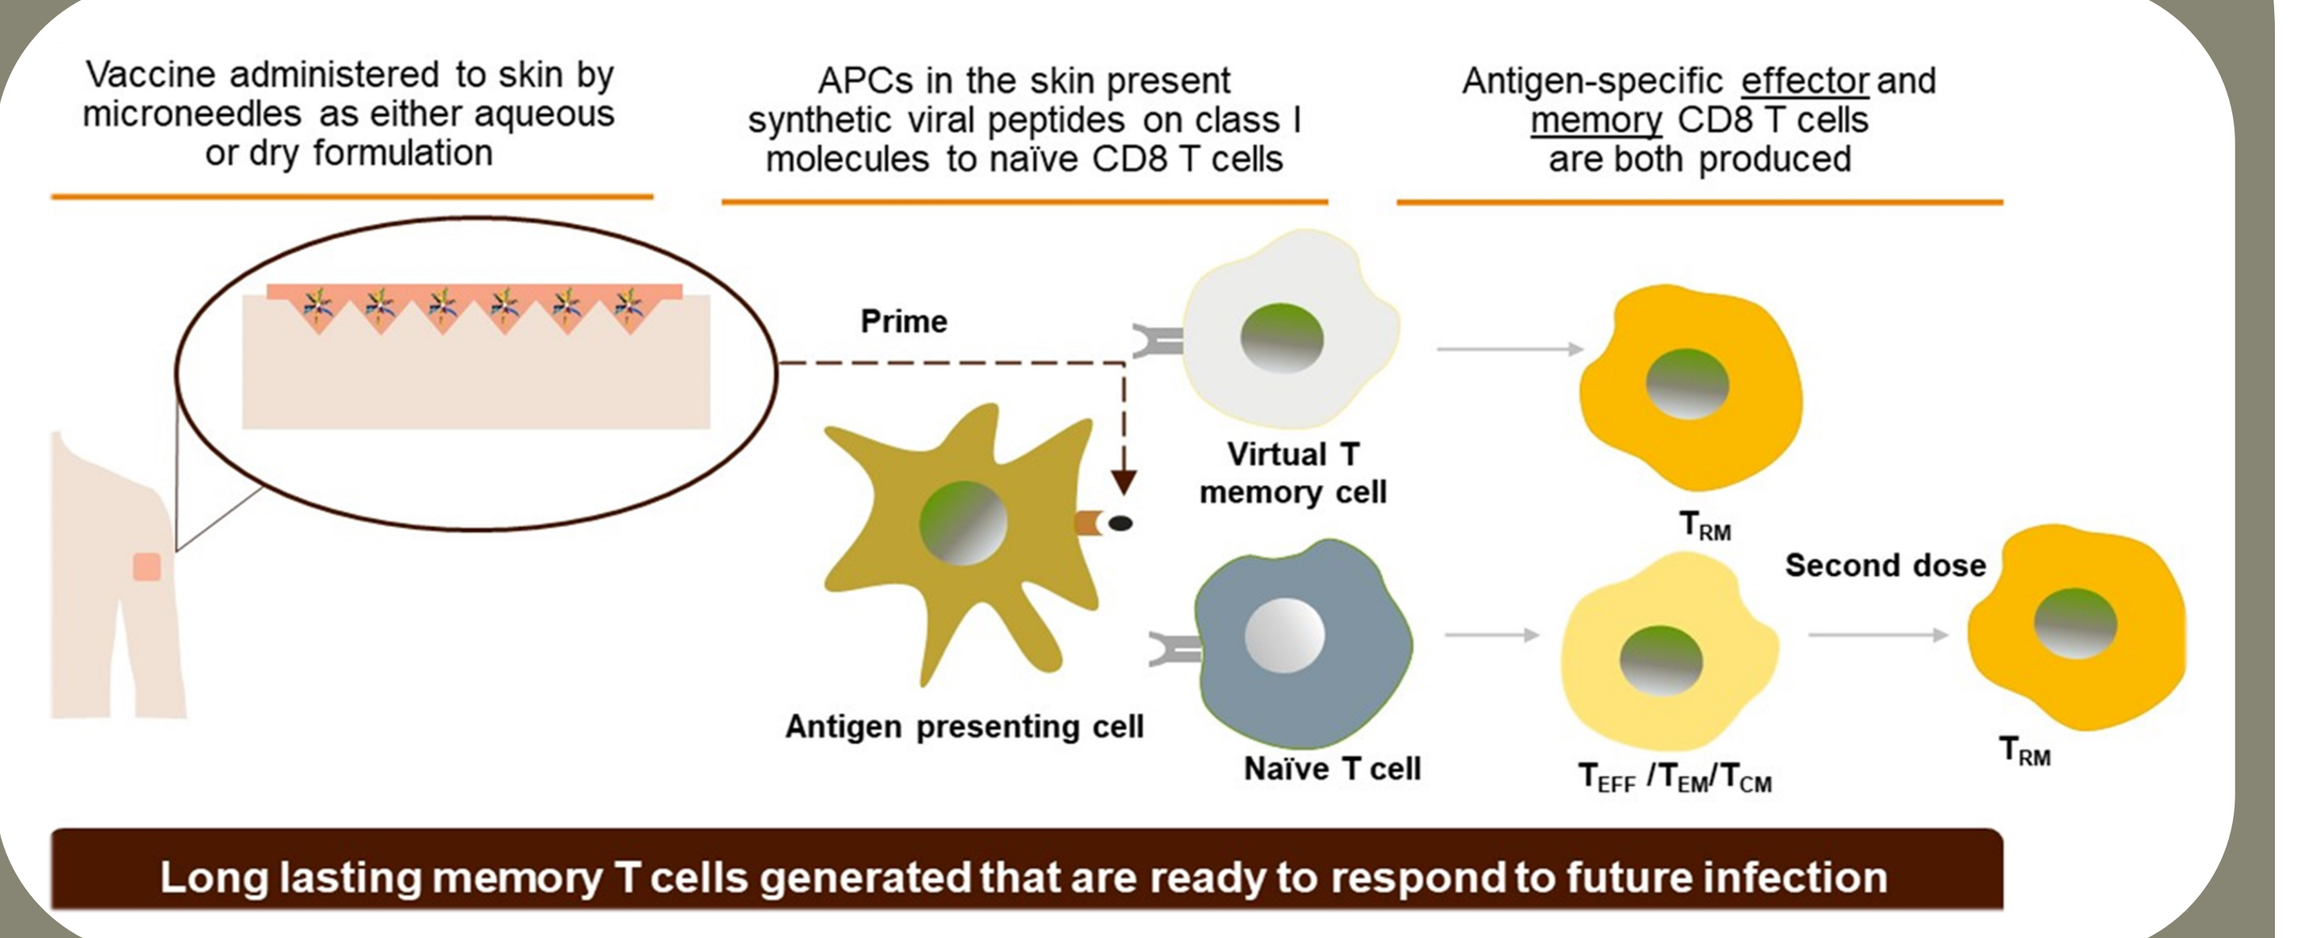

Vaccine administered to skin by microneedles as either aqueous or dry formulation
APCs in the skin present synthetic viral peptides on class I molecules to naïve CD8 T cells
Antigen-specific effector and memory CD8 T cells are both produced
Prime
Virtual T memory cell
Antigen presenting cell
Naïve T cell
TRM
Second dose
TEFF /TEM/TCM
TRM
Long lasting memory T cells generated that are ready to respond to future infection

Secondary vaccination provides necessary "alarm" signals establishing long term T_{RM} niche-immunity

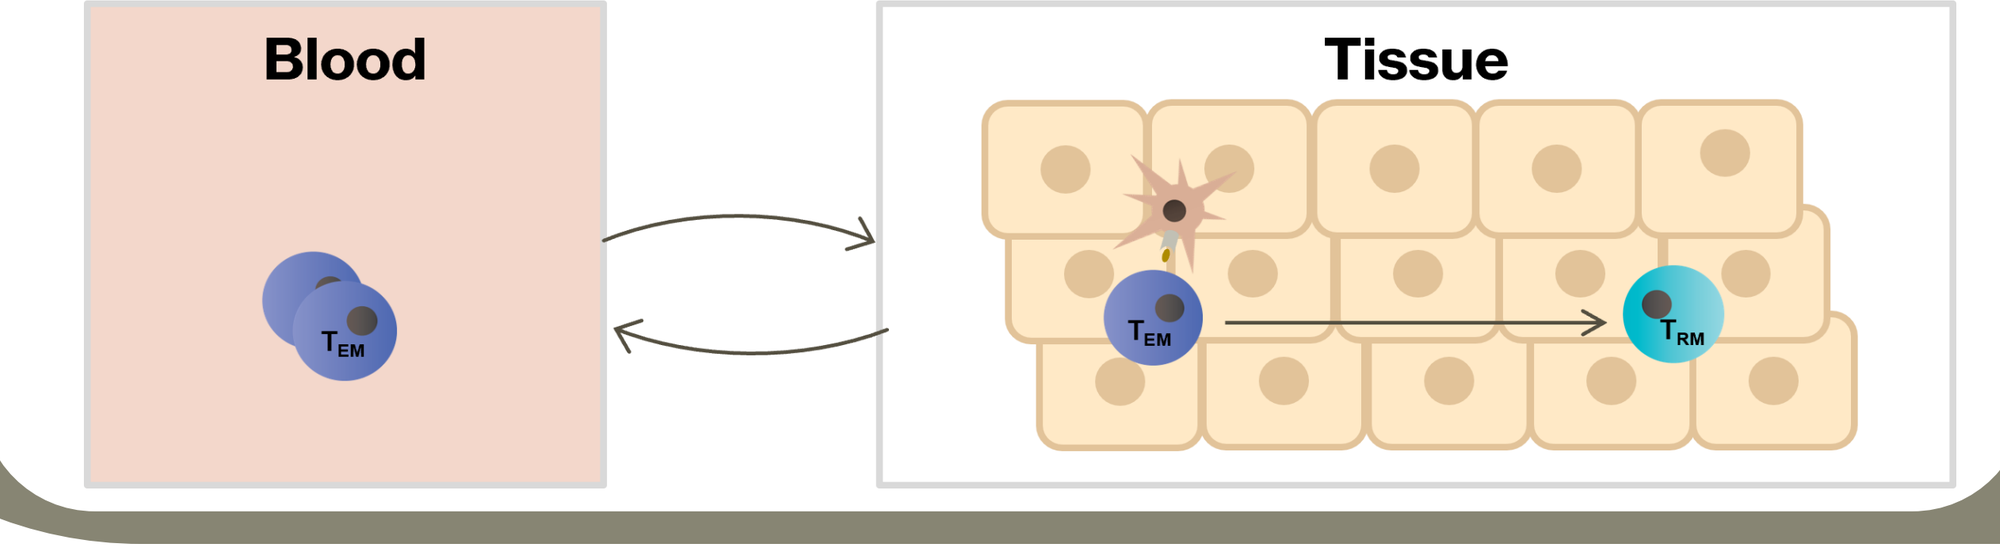

Blood
Tissue
TEM
TEM
TRM

Microneedles mimic natural exposure to a virus increasing immunogenicity of Emergex vaccines

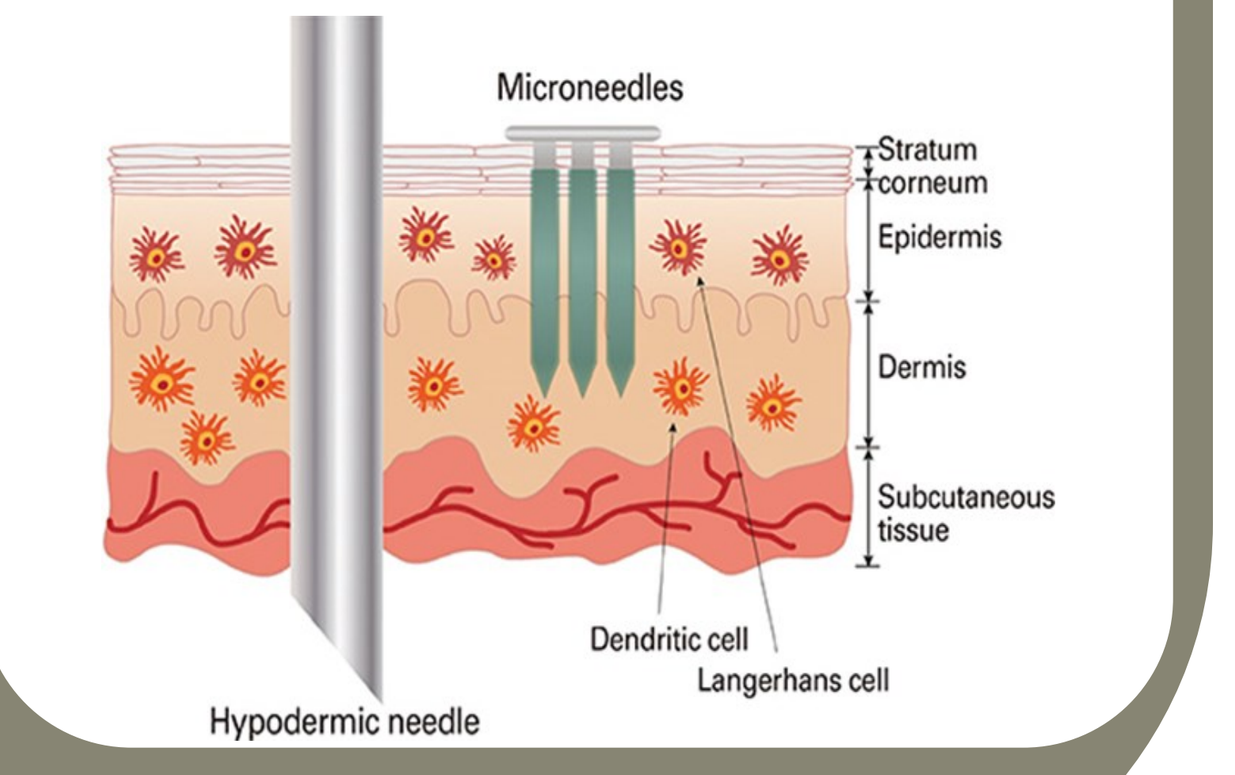

Microneedles
Stratum corneum
Epidermis
Dermis
Subcutaneous tissue
Dendritic cell
Langerhans cell
Hypodermic needle