Characterization of a T cell adaptive vaccine platform to prime a broad and durable immunogenic response against Dengue virus infection

Hager Mohamed* | Aykan Karabudak | Xiaofang Huang
Emergex USA Corporation - Doylestown, PA
*Research Scientist; h.mohamed@emergexvaccines.com

Background Information

- Dengue hemorrhagic fever, caused by the Dengue virus (DENV), is a mosquito-borne disease affecting millions of people worldwide, with cases increasing to over 8-fold in the last 20 years.
- Current vaccination approaches focus on eliciting an antibody response against DENV, which may cause antibody-mediated enhancement (ADE).
- **Emergex Vaccines' T cell** adaptive vaccine platform is intended to evoke cellular immune responses similar to those initiated in natural viral infection.
- Emergex's fully synthetic **Dengue T cell priming (Dengue TCP) vaccine** utilizes multiple virus-specific peptides attached to a gold nanoparticle.
- Emergex's Dengue TCP vaccine can be administered intradermally by a **microneedle delivery system** for optimal uptake by antigen presenting cells, which subsequently display the virus-specific peptides onto their Major Histocompatibility Class I molecules (MHC I) to activate **CD8+ T cells** (*Fig 1*).

The T cell adaptive immune response generated by Emergex vaccines

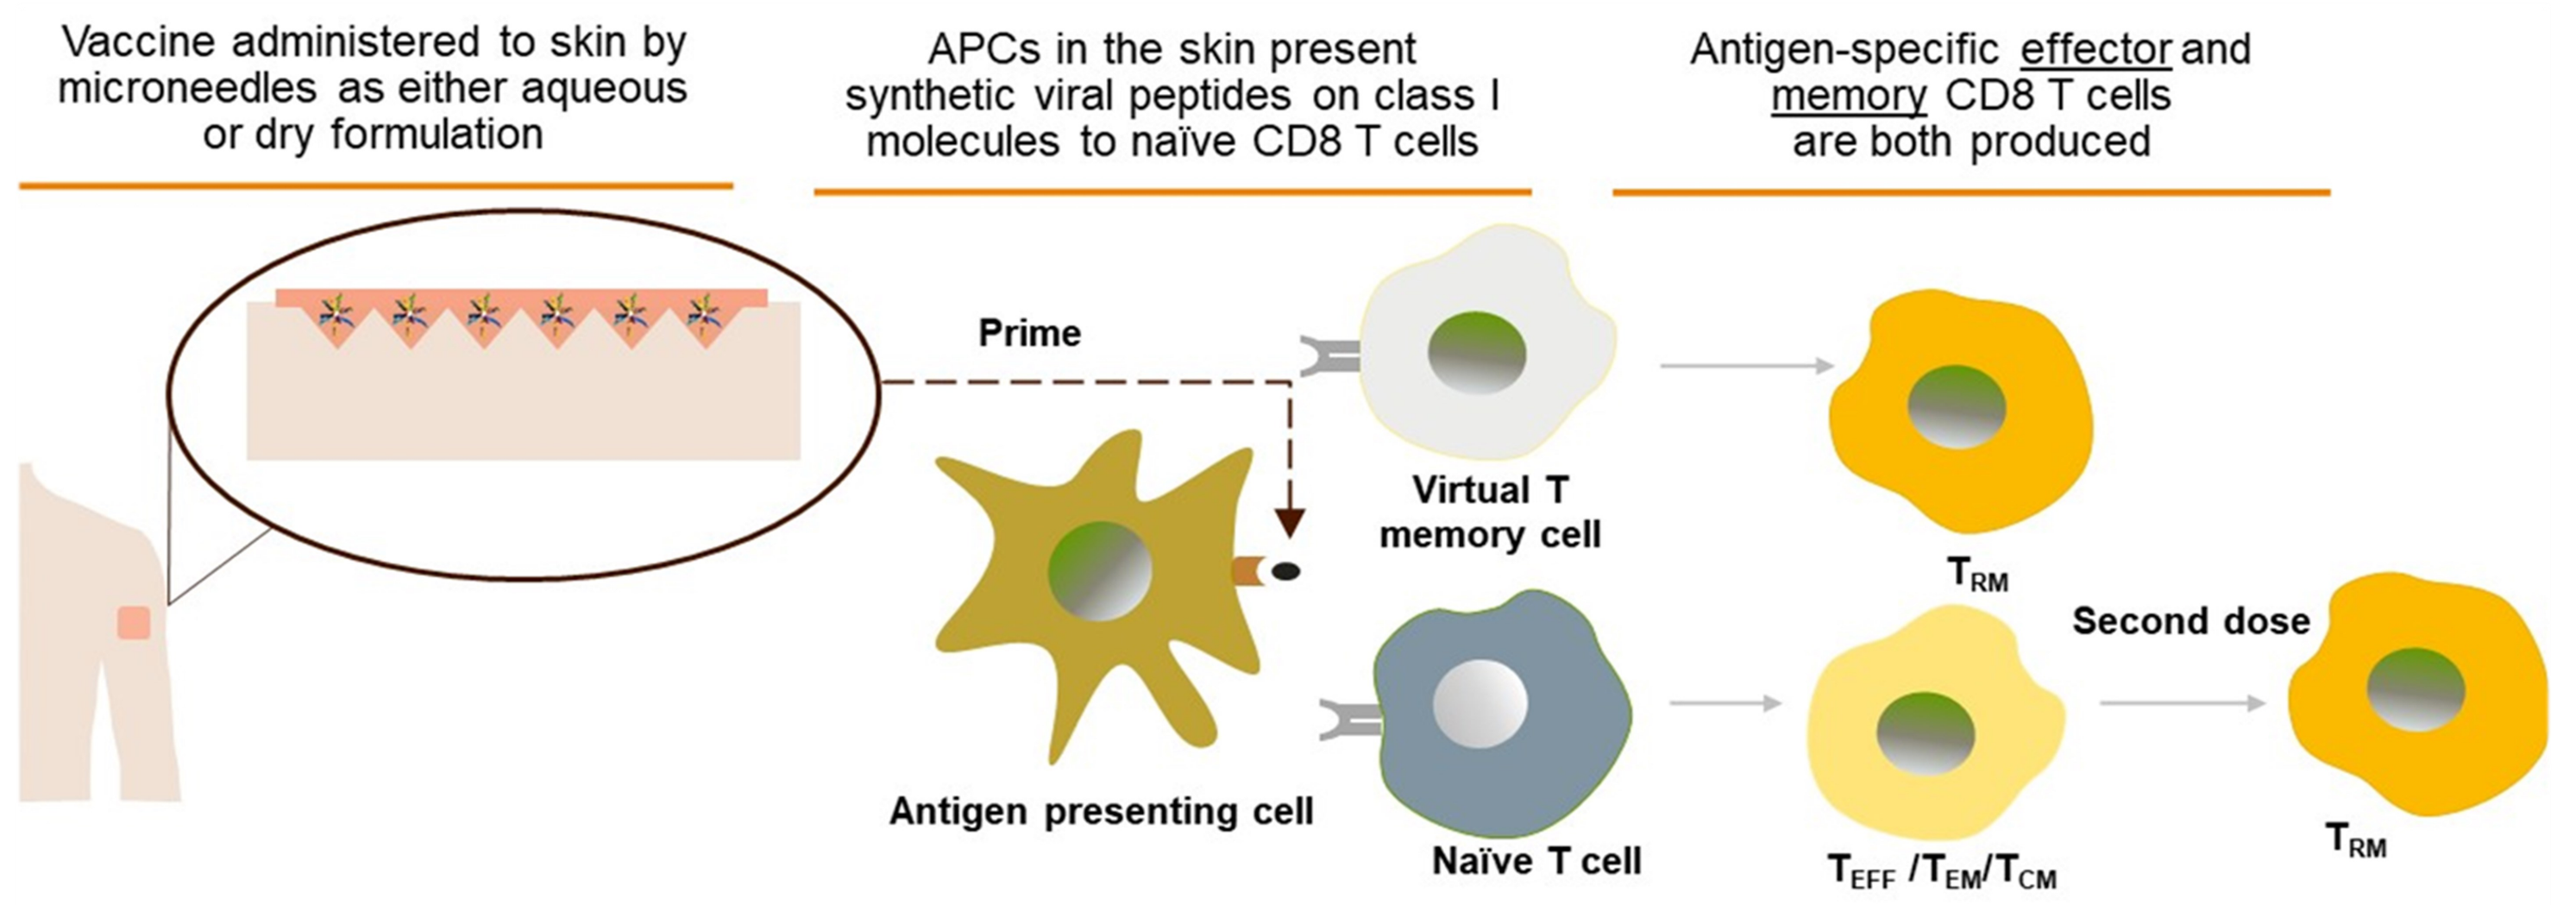
Vaccine administered to skin by microneedles as either aqueous or dry formulation
APCs in the skin present synthetic viral peptides on class I molecules to naïve CD8 T cells
Antigen-specific effector and memory CD8 T cells are both produced
Prime
Virtual T memory cell
T_{RM}
Second dose
Antigen presenting cell
Naïve T cell
T_{EFF} /T_{EM}/T_{CM}
T_{RM}

Figure 1. The initial Emergex Dengue TCP vaccination stimulates dendritic cells (DCs) in the skin to process and present vaccine peptides on MHC class I molecules to naïve T cells in the draining lymph nodes. This causes expansion of T effector cells (TEFF) and T memory cells that can migrate to the tissues. Upon a second vaccination, distal DCs can present antigen to circulating and marginating T memory cells which can differentiate into mature long-term tissue resident memory cells (TRM) in response to antigen.

Emergex Dengue TCP Vaccine Design

DENV Infection of HepG2 and JY cells

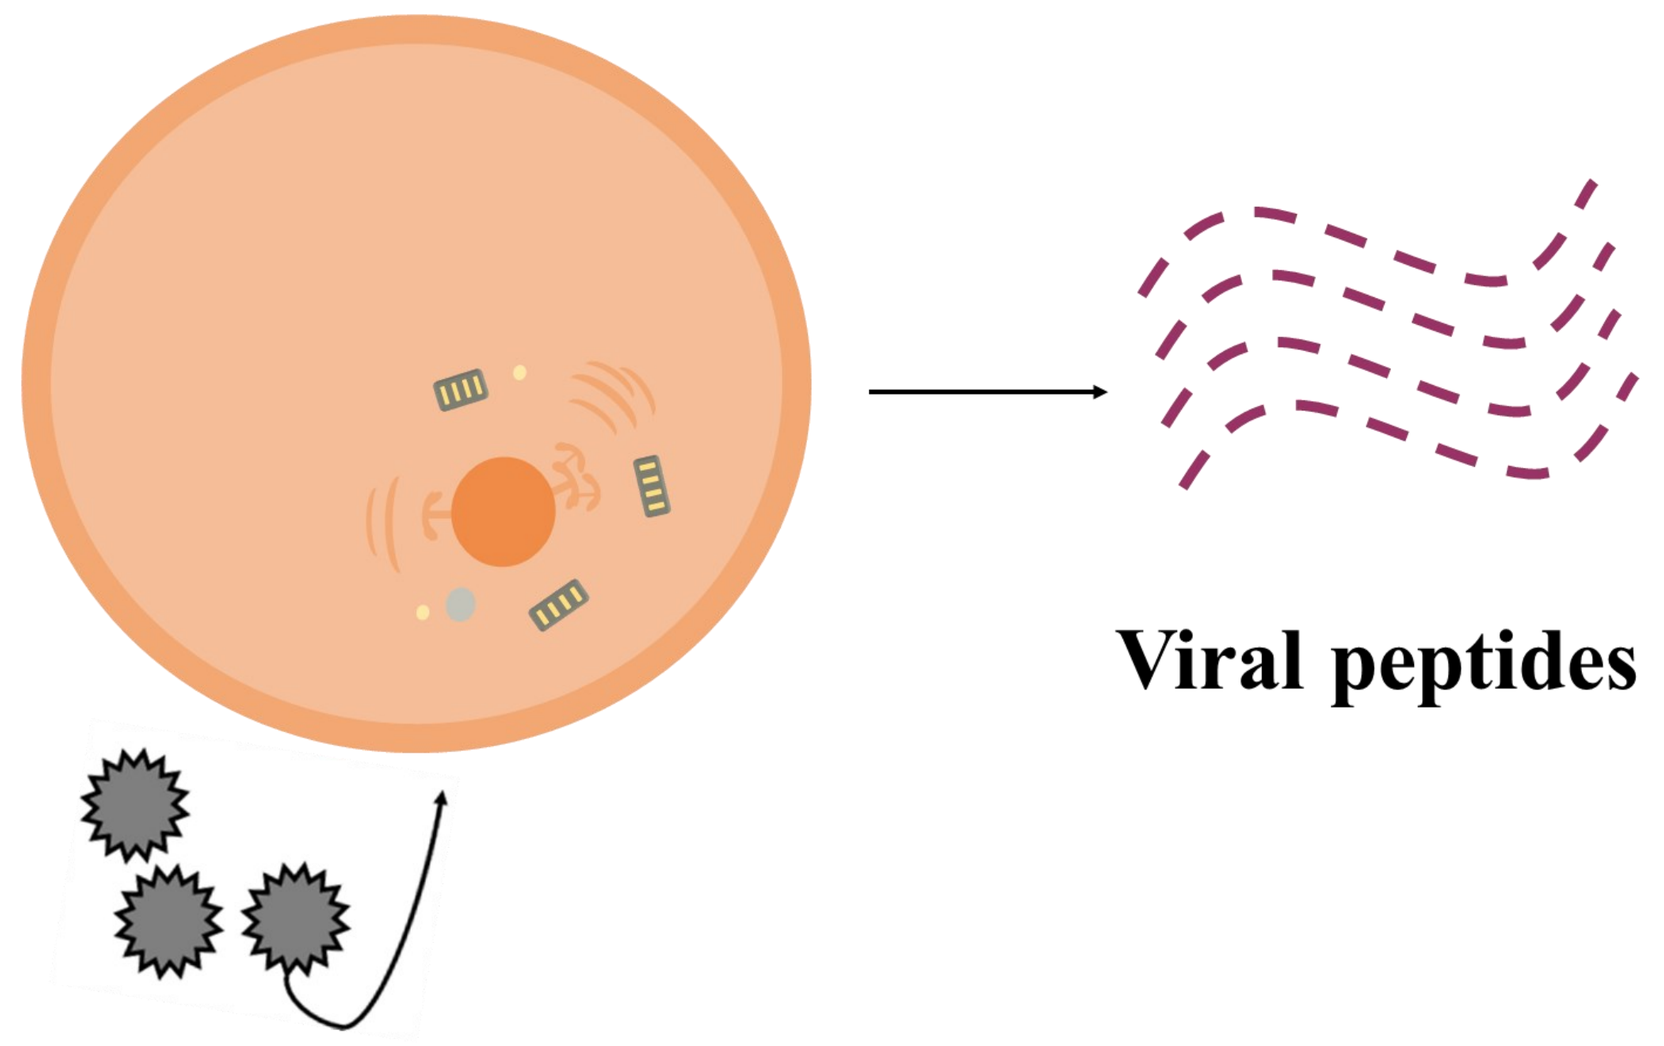
Viral peptides

Ligandome Library Generation by Immunoproteomics

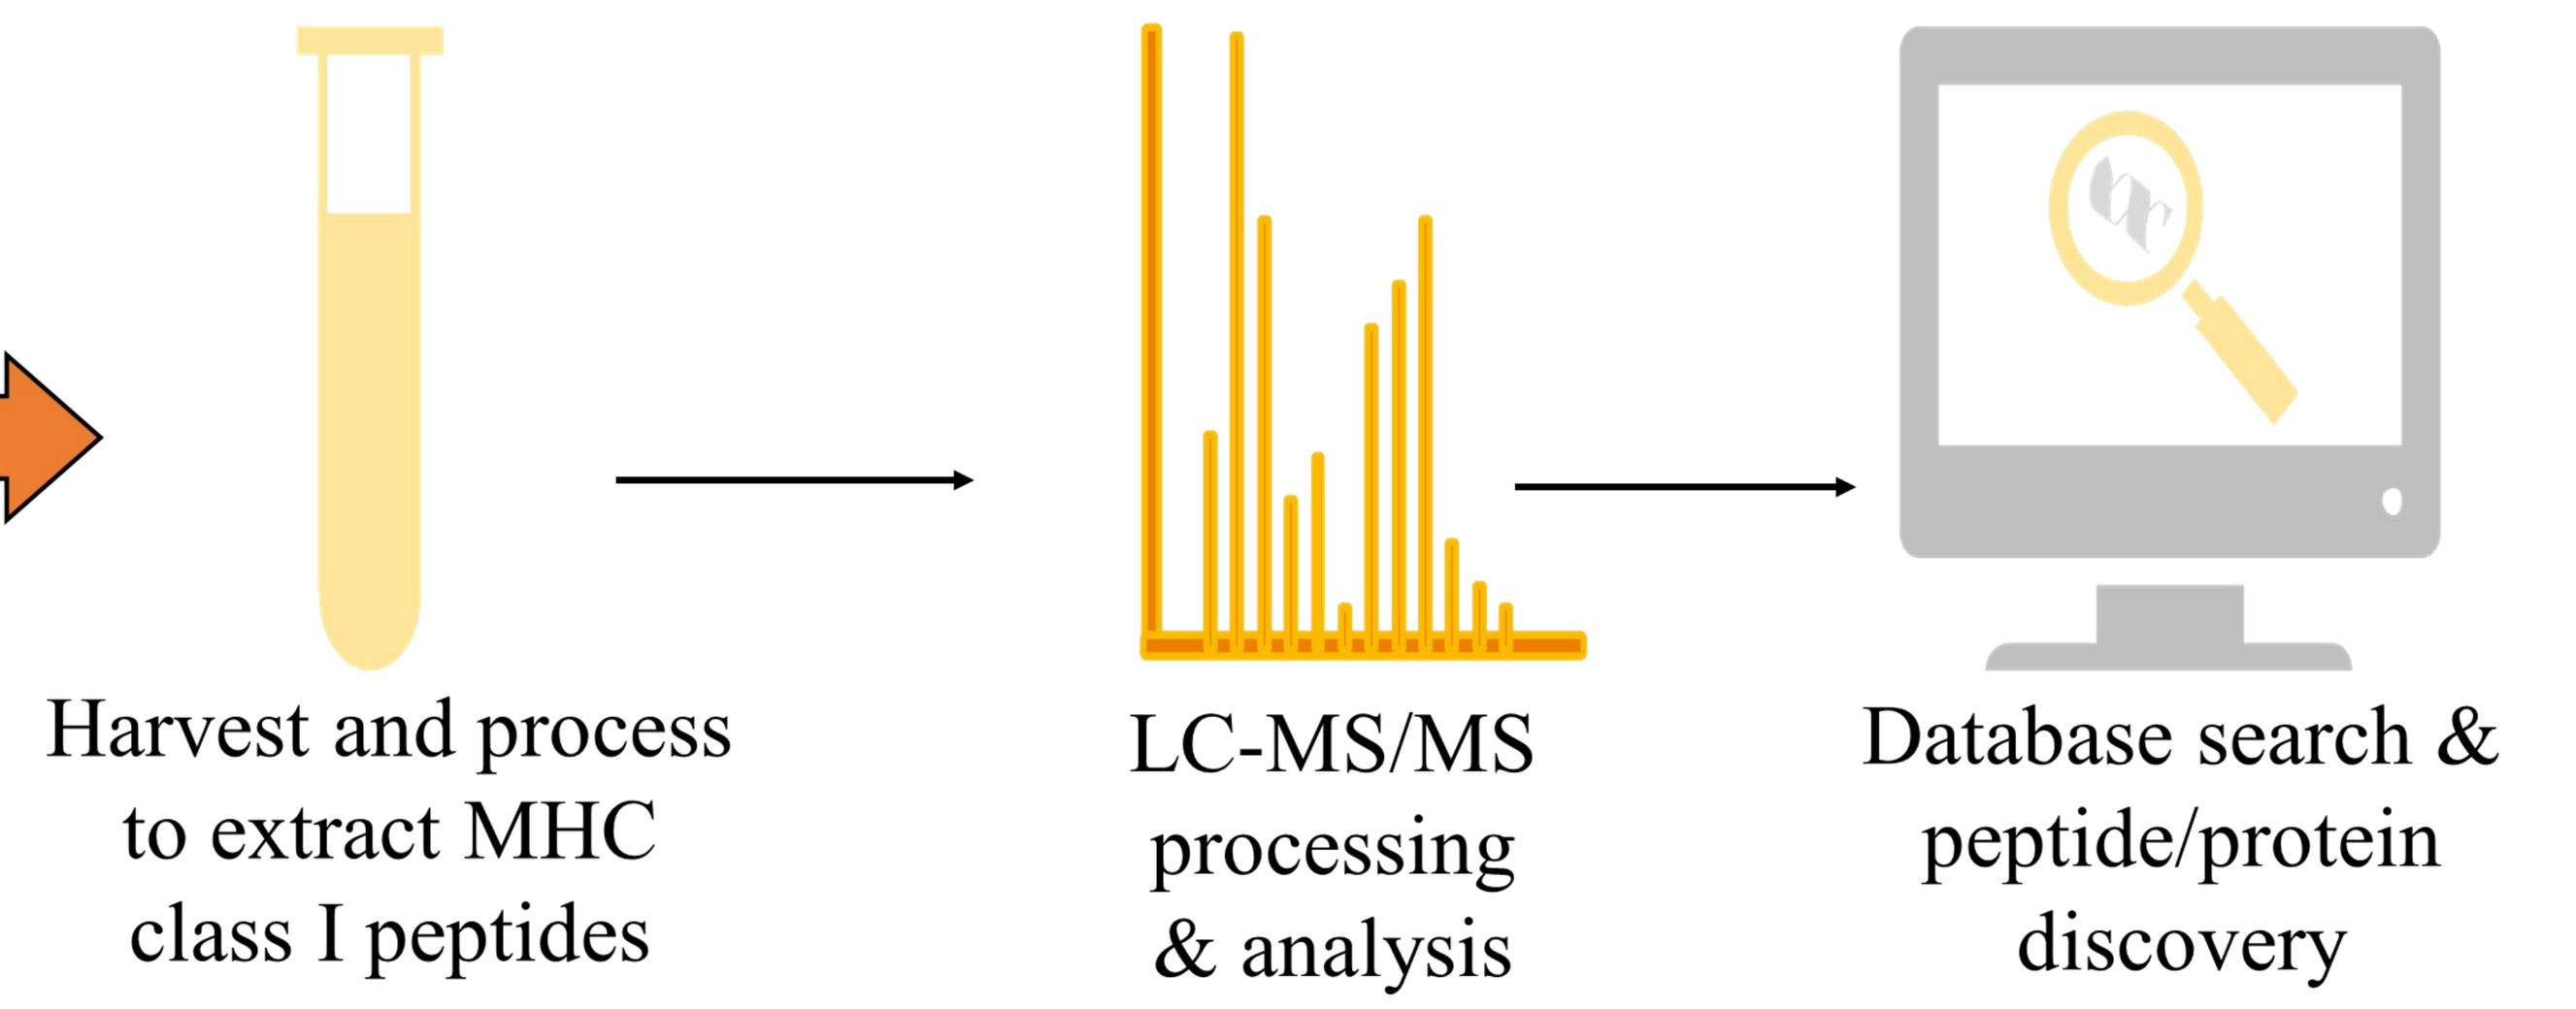
Harvest and process to extract MHC class I peptides
LC-MS/MS processing & analysis
Database search & peptide/protein discovery

Microneedle Array Patch Delivery

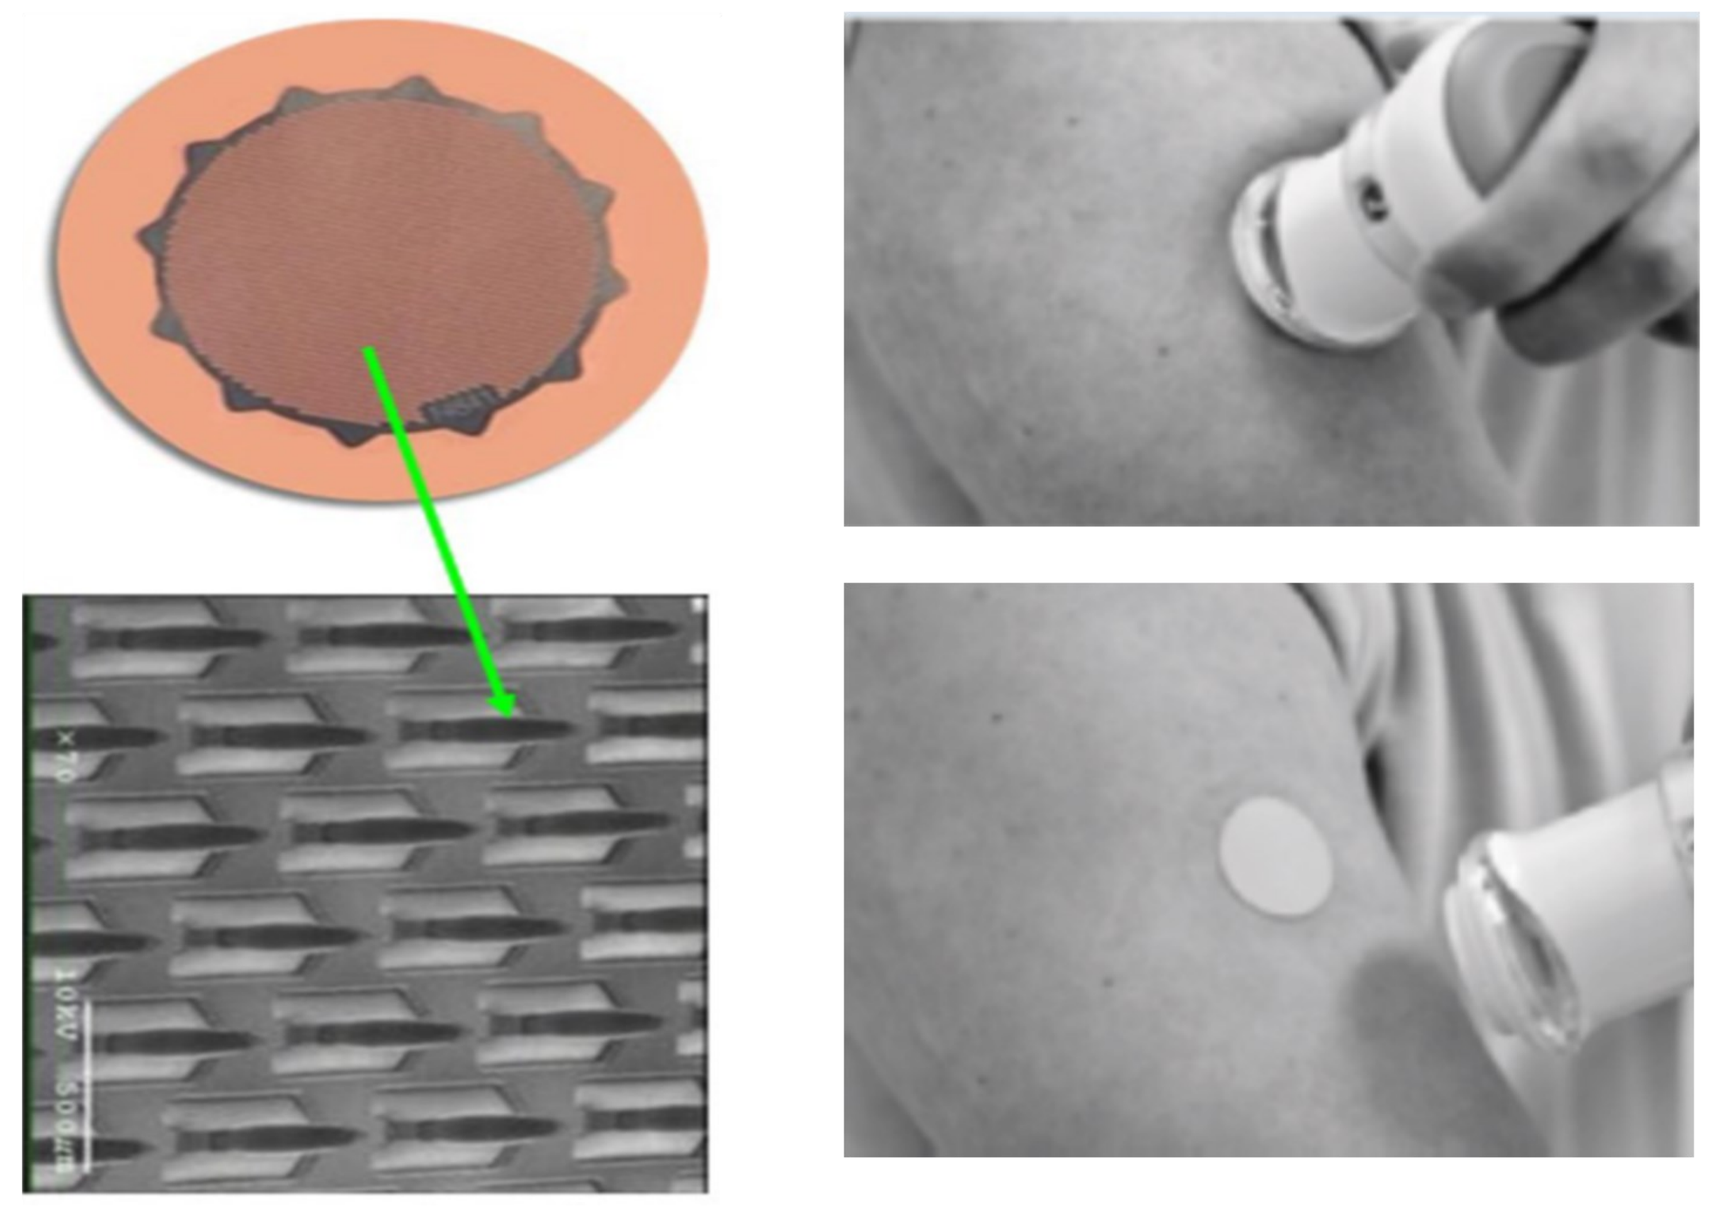

Formulation with Gold Nanoparticle Vehicle for Delivery

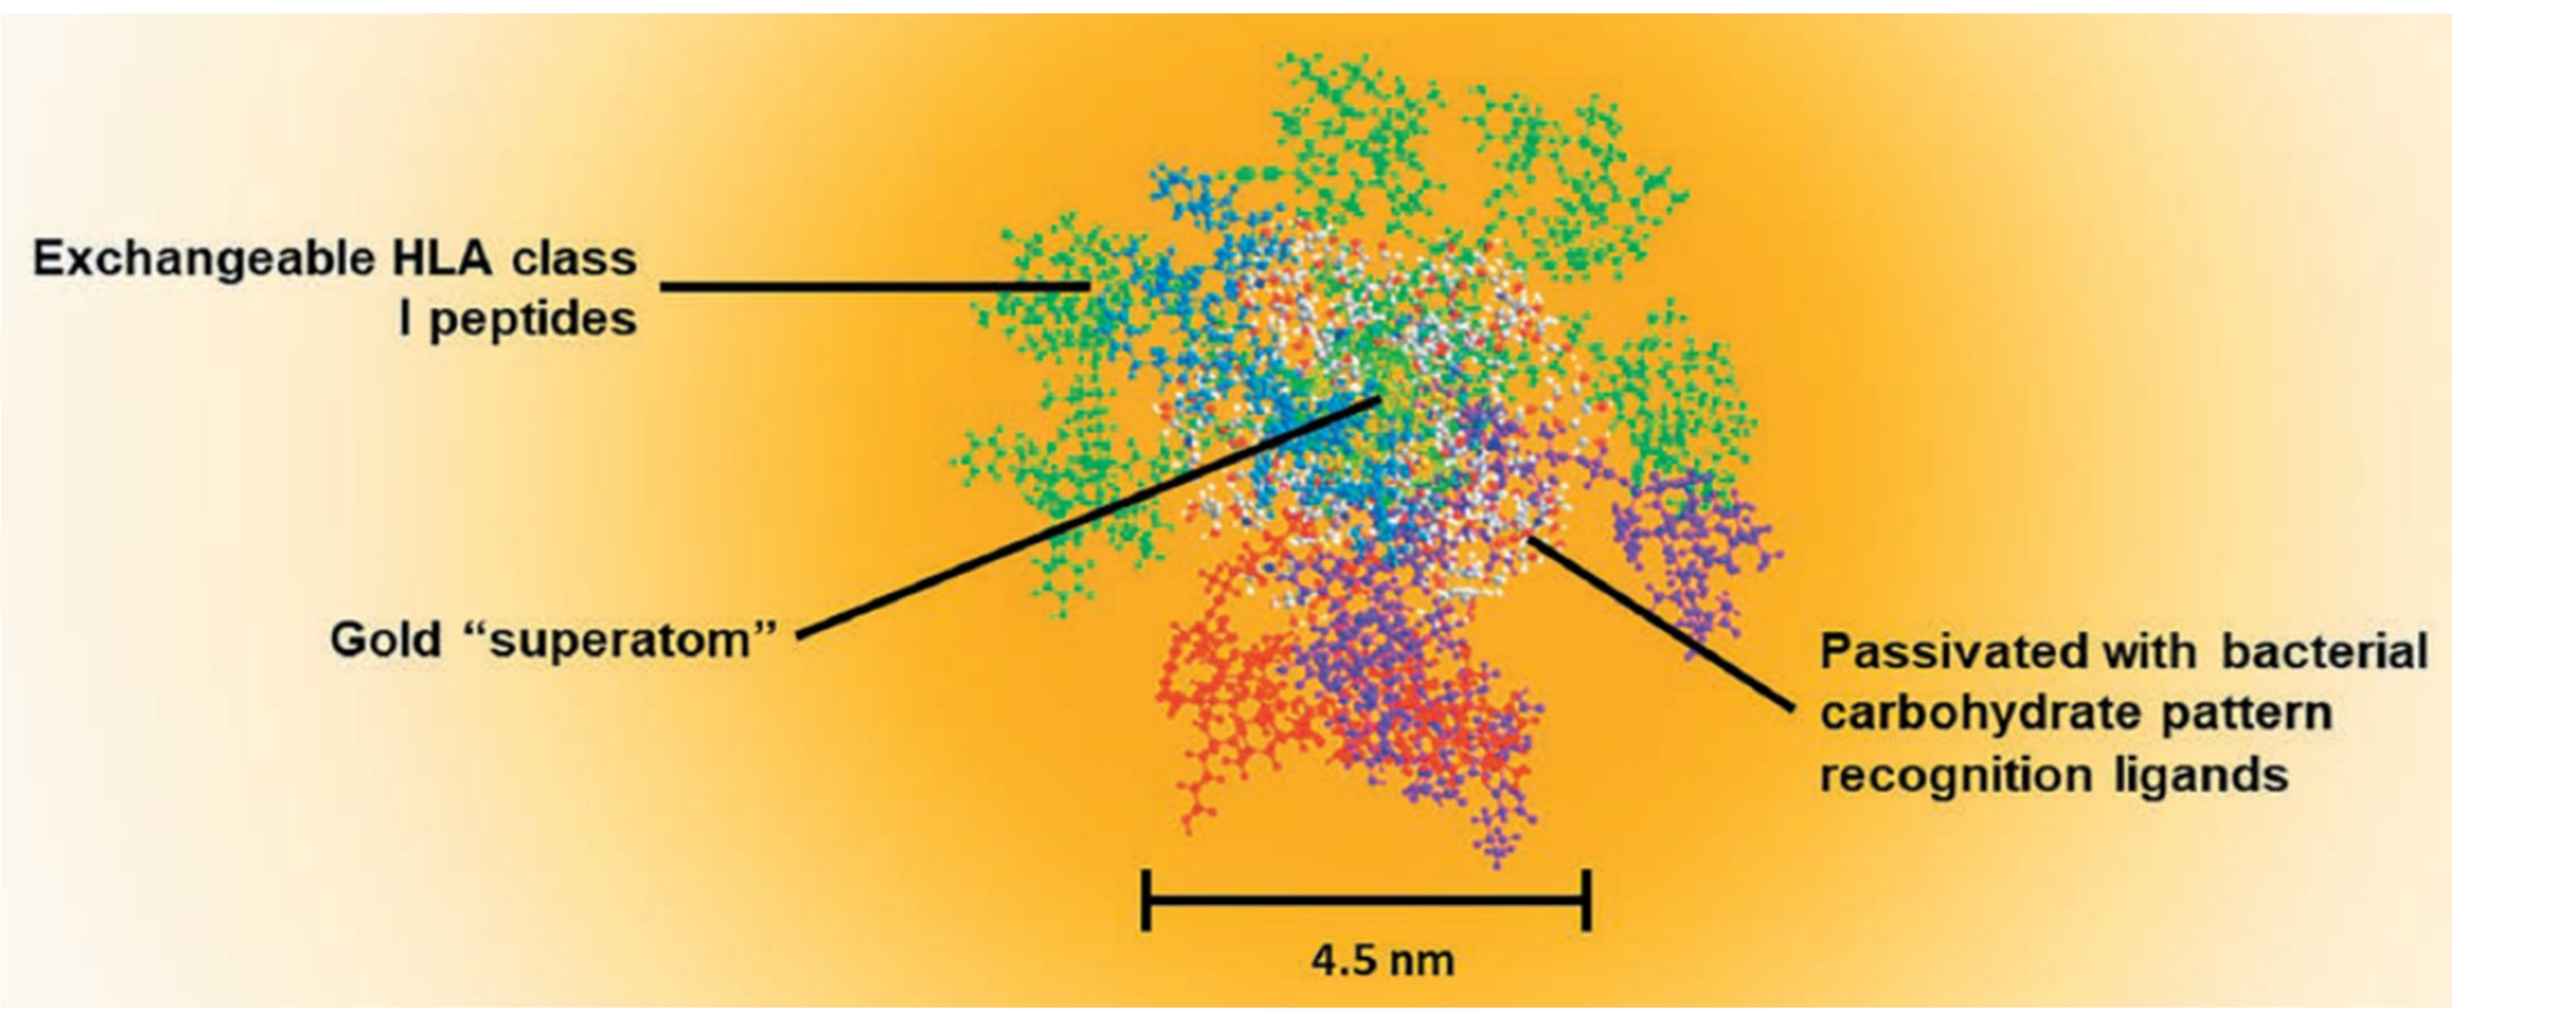
Exchangeable HLA class I peptides
Gold "superatom"
Passivated with bacterial carbohydrate pattern recognition ligands
4.5 nm

Figure 2. The fully synthetic Dengue TCP vaccine is composed of Dengue virus specific peptides with a gold nanoparticle vehicle. MHC class I restricted, virus specific peptides were harvested from cell models (mainly HepG2 and JY cells) infected with Dengue virus; then vaccine candidates were identified using an immunoproteomic method. Candidate peptides were initially characterized *in vitro* for immunogenicity by co-culturing T cells from a healthy PBMC donor expanded through peptide-stimulation with virally infected antigen presenting cells (HepG2 cells). Dengue specific candidate peptides were then conjugated with a gold nanoparticle vehicle for *in vivo* characterization and studies.

Results

Peptide candidate selection based on LC-MS/MS processing & analysis

Peptide No.	Protein Source	HLA motif	Infected cell type
P1	NS2A	A2/24	HepG2
P2	NS4B	A2	Dendritic cell
P3	NS5	A2/24	HepG2
P4	NS5	A2/24	JY
P5	Polyprotein	A2/24	HepG2
P6	NS2A	A2/24	HepG2

Table 1. List of MHC class I restricted peptides candidates for assessment, along with associated DENV protein, HLA type (A2 or A24), and cell model used for infection and peptide discovery.

In vitro characterization of peptide immunogenicity

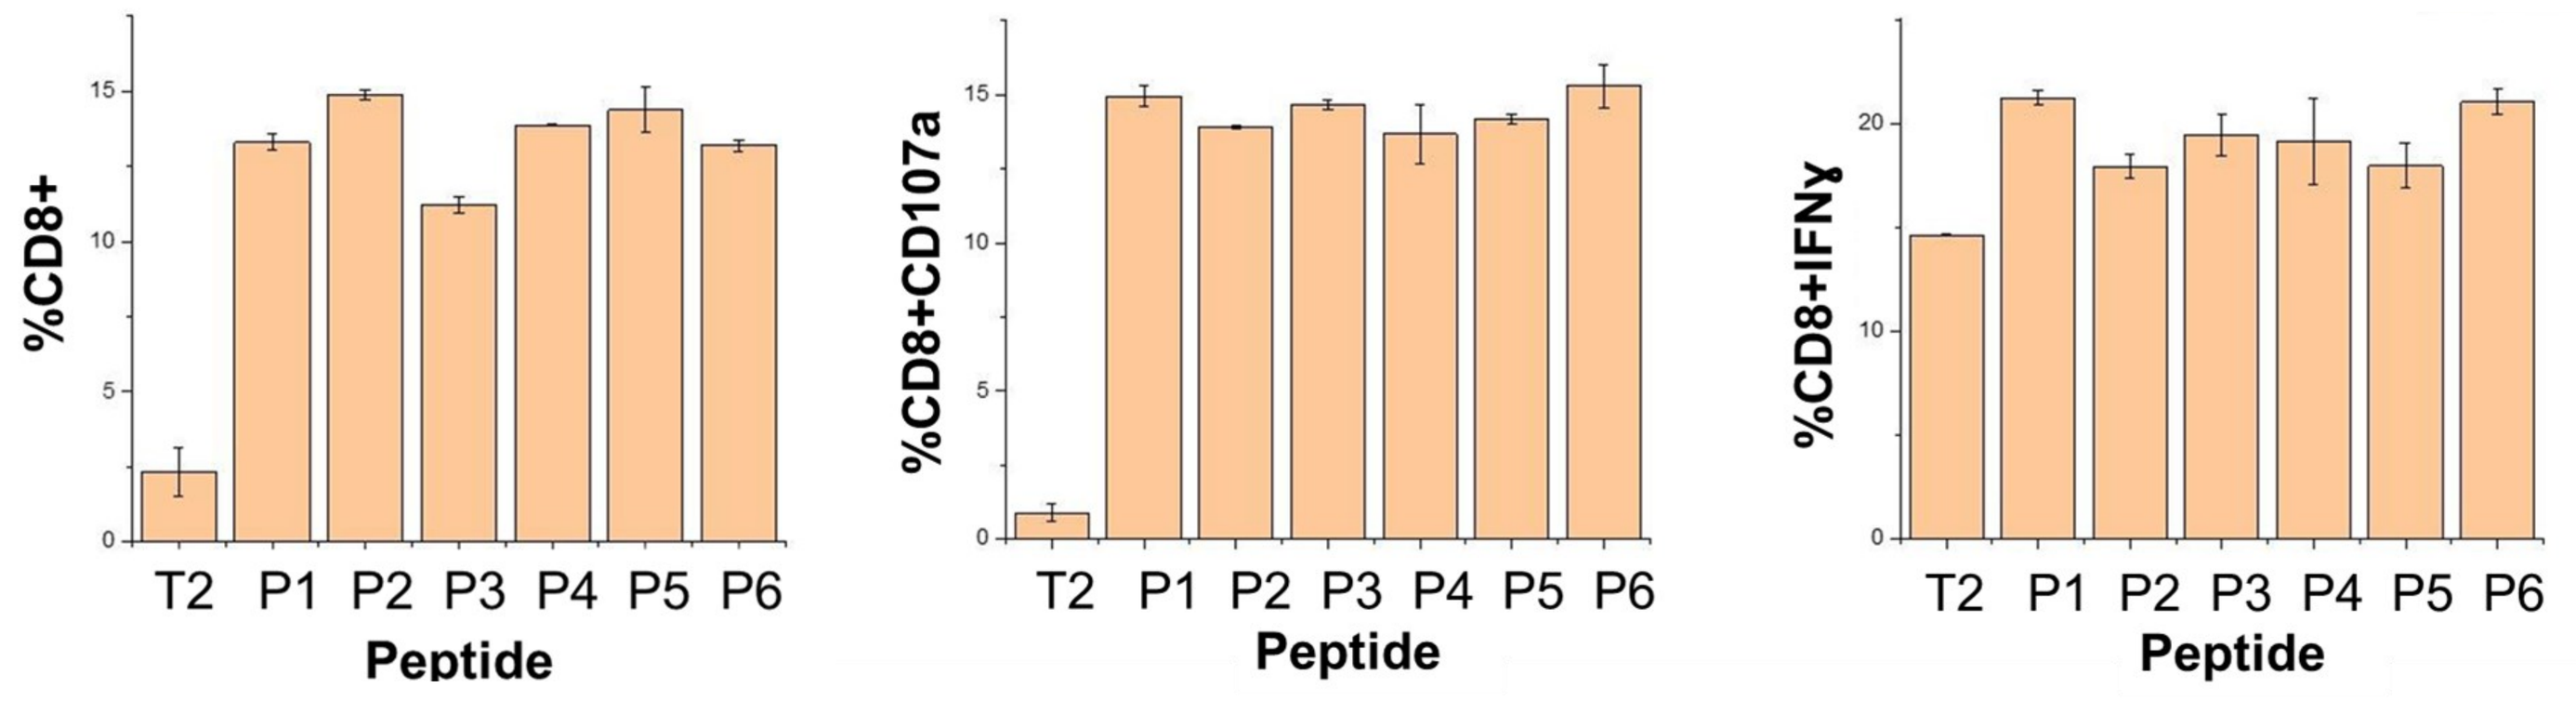
%CD8+
%CD8+CD107a
%CD8+IFNɣ
T2 P1 P2 P3 P4 P5 P6
Peptide

Figure 3. Antigen-specific response of CD8+ T cells characterized in PBMCs from a healthy human HLA-A2+ donor, following stimulation with individual MHC class I restricted peptide epitopes detailed in *Table 1*. PBMCs were co-cultured with Tap deficient T2 cells alone (T2) or T2 cells stimulated with individual peptides (*Table 1*, P1-P6). CD8+ T cell activation was assessed through the expression of the degranulation marker CD107a and the antiviral cytokine IFNɣ, measured by flow cytometry.

Evaluation of the T cell response to Dengue TCP vaccine following immunization of HLA-A2 transgenic mice

Experimental overview

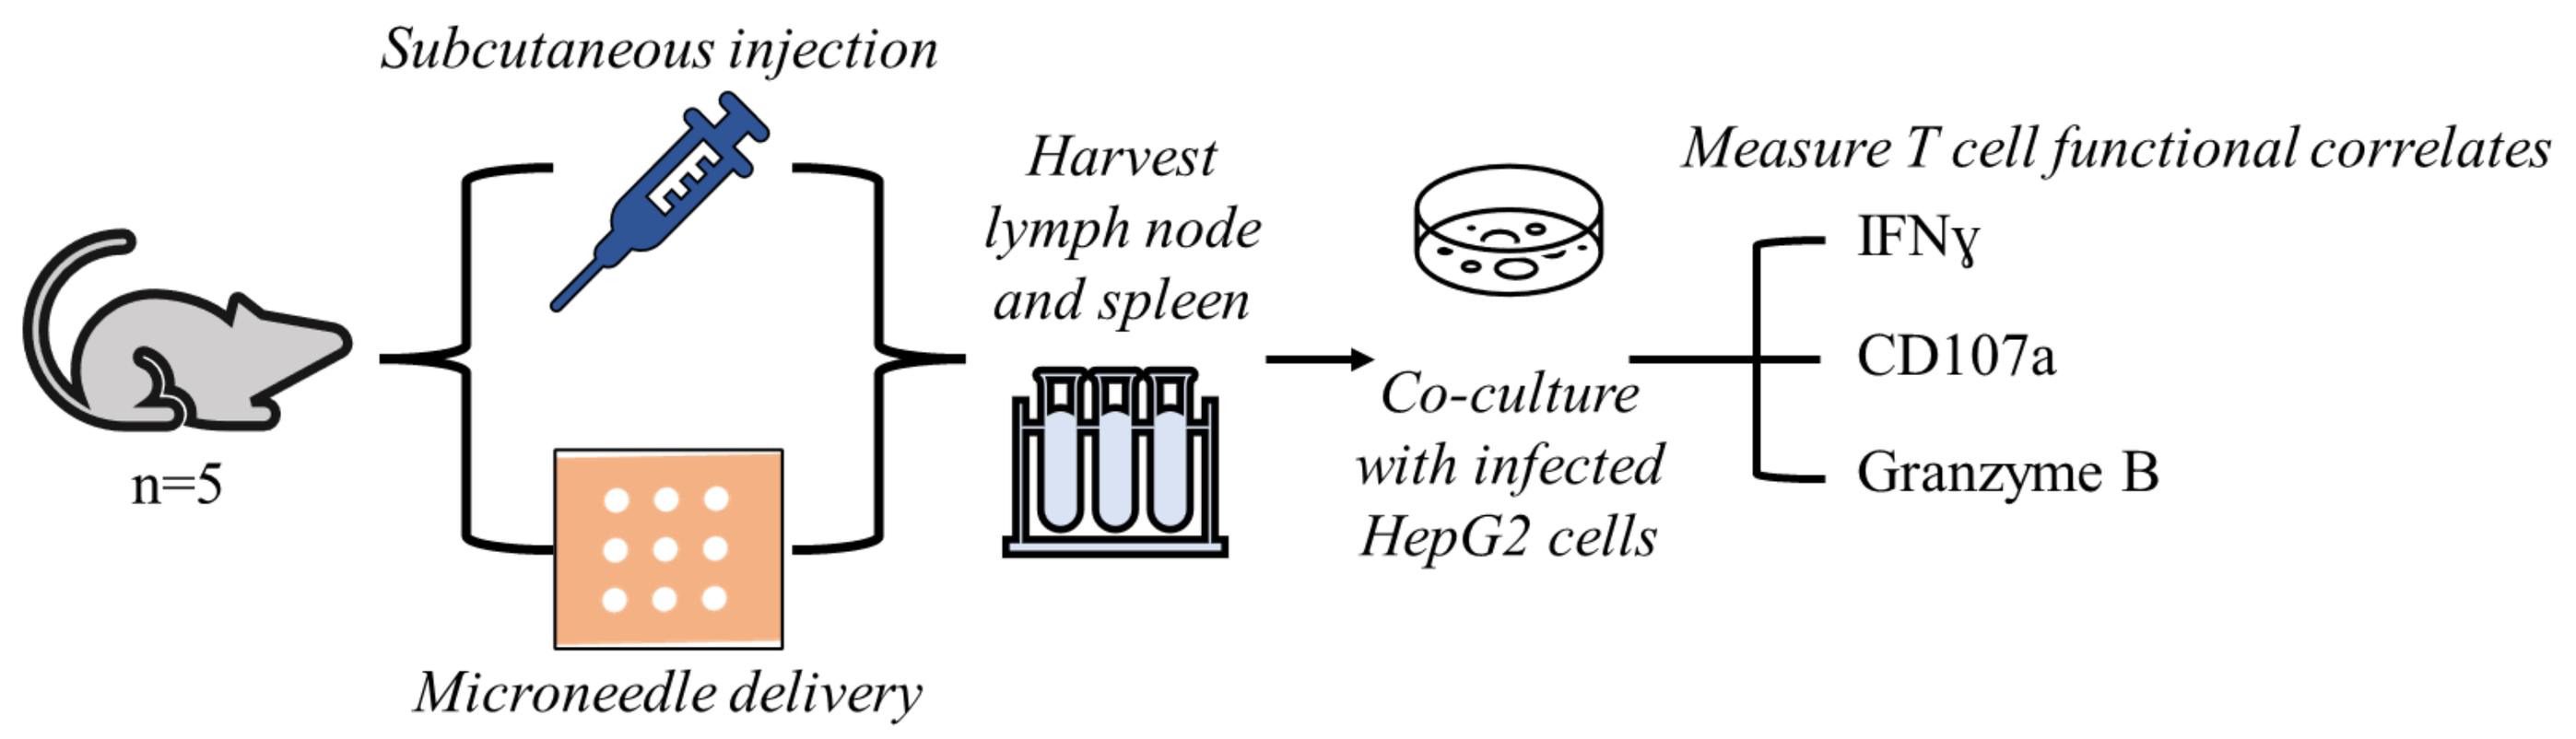
Subcutaneous injection
n=5
Microneedle delivery
Harvest lymph node and spleen
Co-culture with infected HepG2 cells
Measure T cell functional correlates
IFNɣ
CD107a
Granzyme B

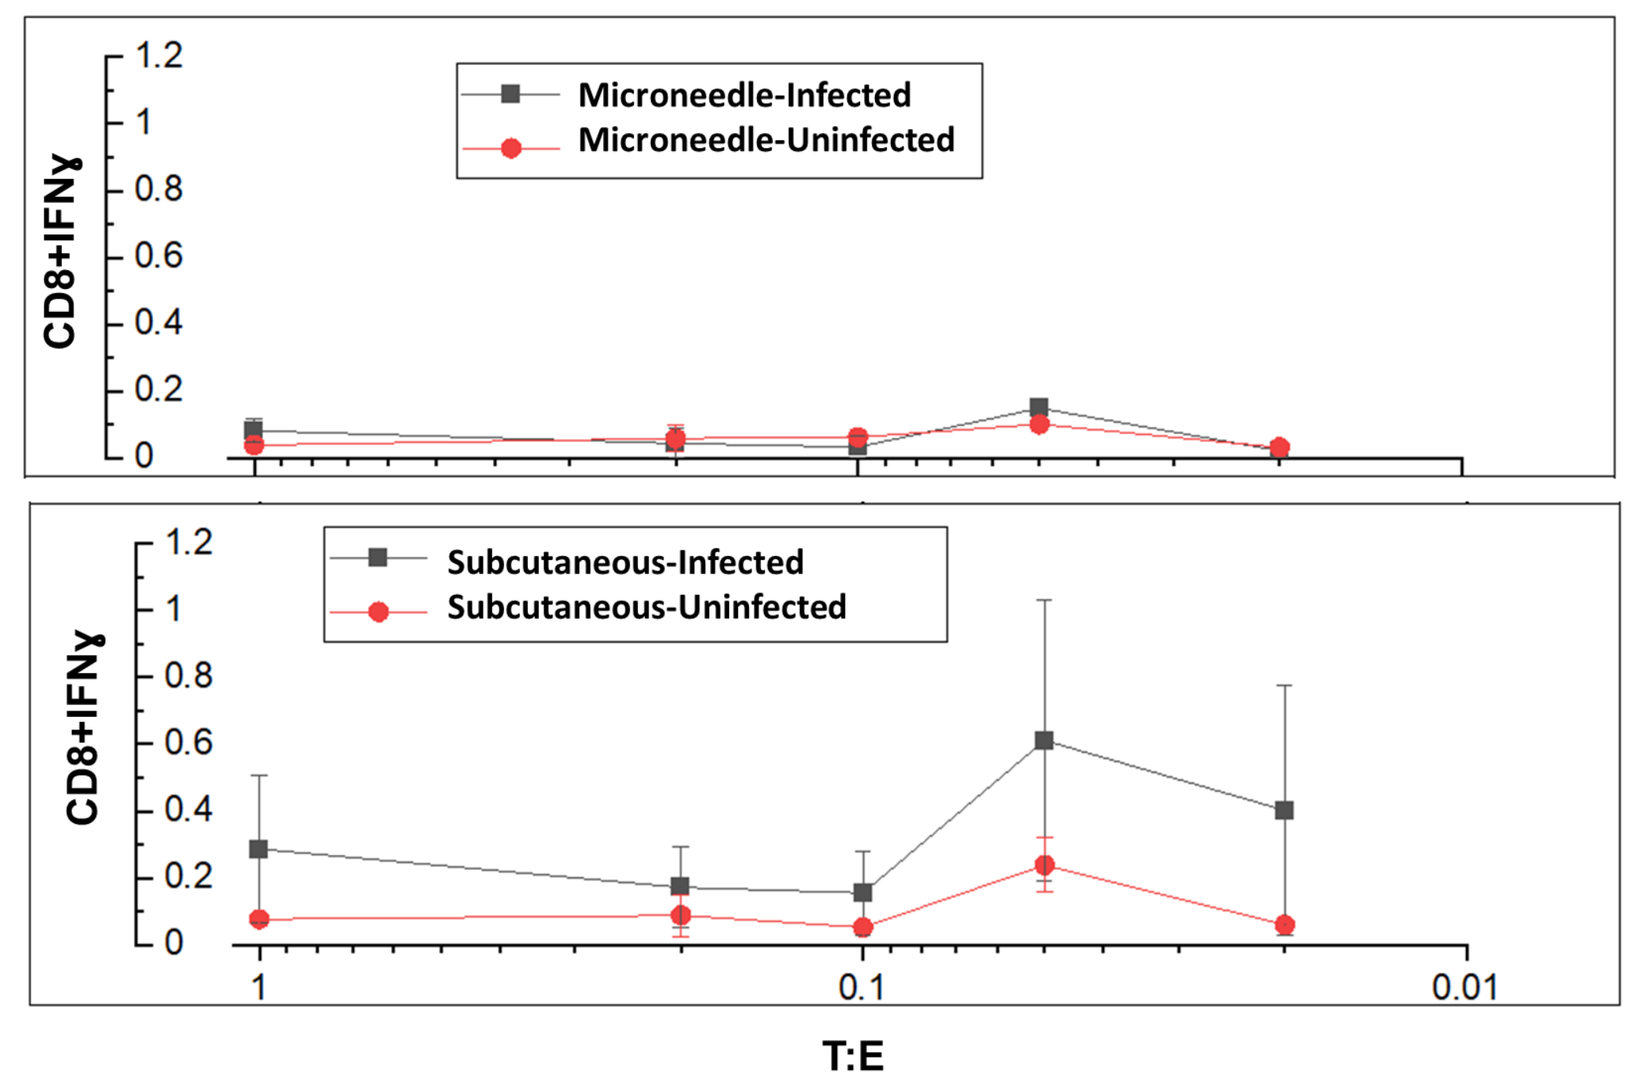
Microneedle-Infected
Microneedle-Uninfected
Subcutaneous-Infected
Subcutaneous-Uninfected
CD8+IFNɣ
T:E

Figure 4. Proportion of CD8+ IFNγ+ cells in inguinal lymph nodes isolated from transgenic HLA-A2 mice immunized with the Dengue TCP vaccine formulation using different injection methods (subcutaneous or microneedle patch). Cells isolated from the spleen were co-cultured with target cells (HepG2), which were either uninfected or infected with Dengue virus, at different target to effector (T:E) cell ratios. IFNɣ expression was evaluated using flow cytometry.

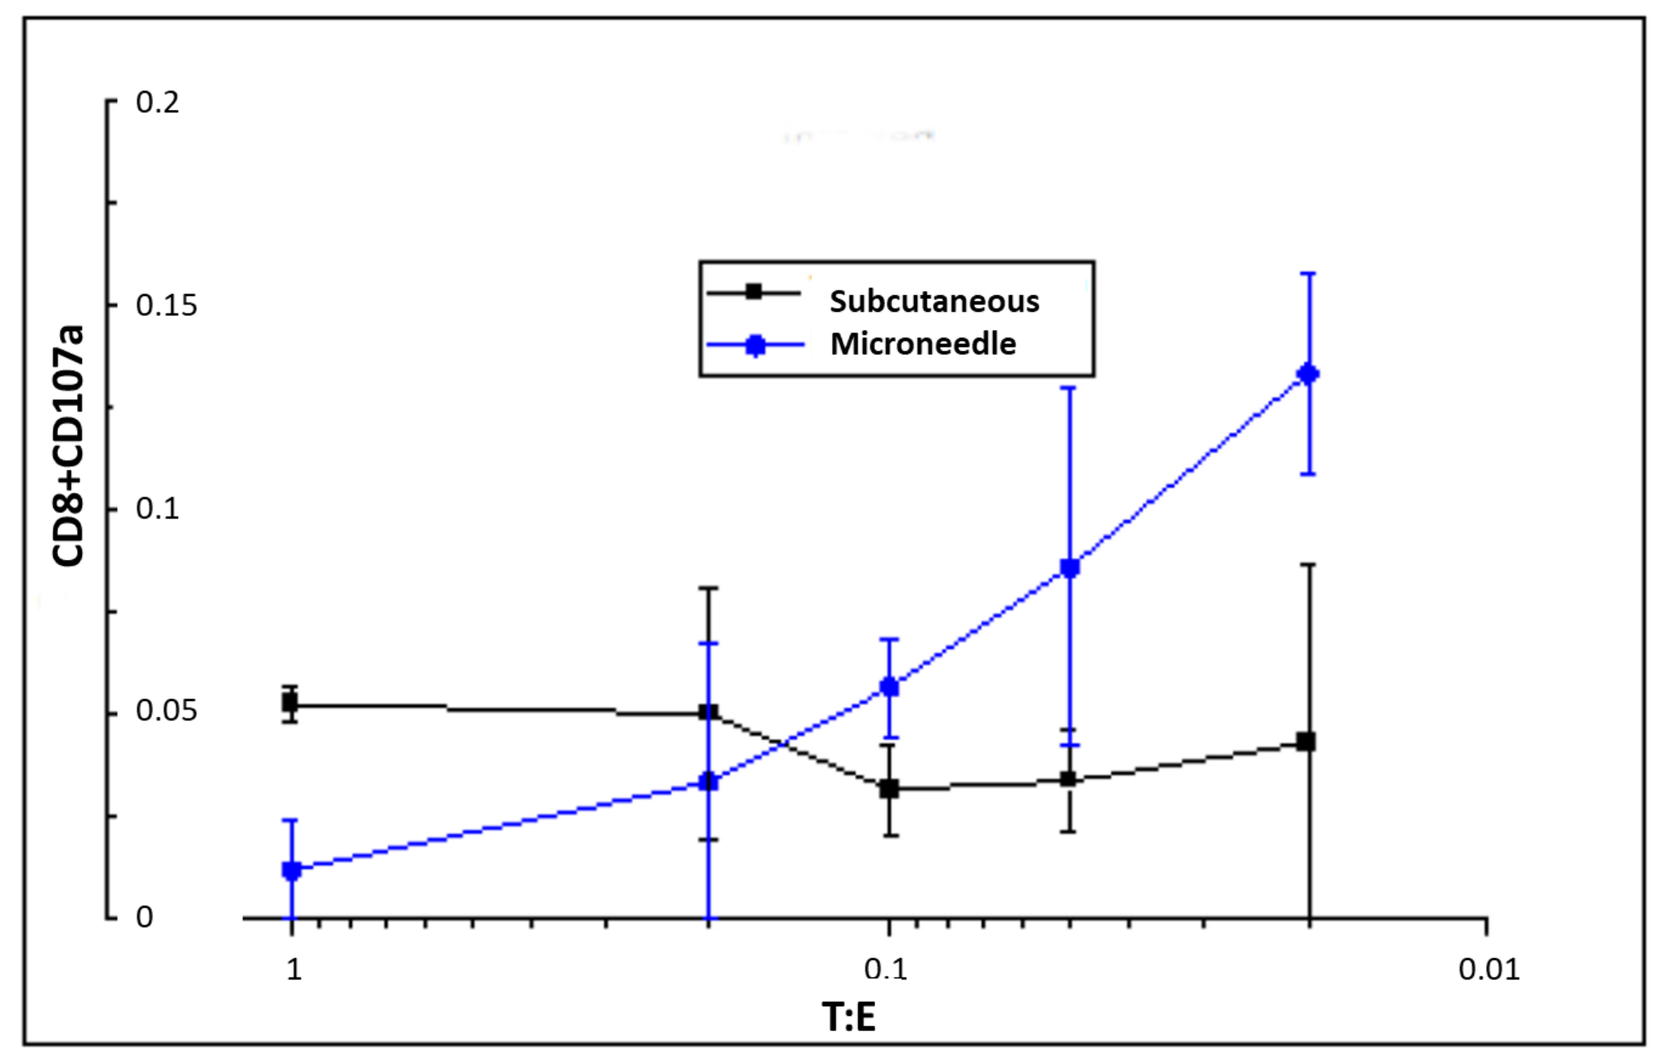
Subcutaneous
Microneedle
CD8+CD107a
T:E

Figure 5. Proportion of CD8+ CD107a+ cells in inguinal lymph nodes isolated from transgenic HLA-A2 mice immunized with the Dengue TCP vaccine formulation using different injection methods and then co-cultured with Dengue-infected target HepG2 cells at different T:E ratios.

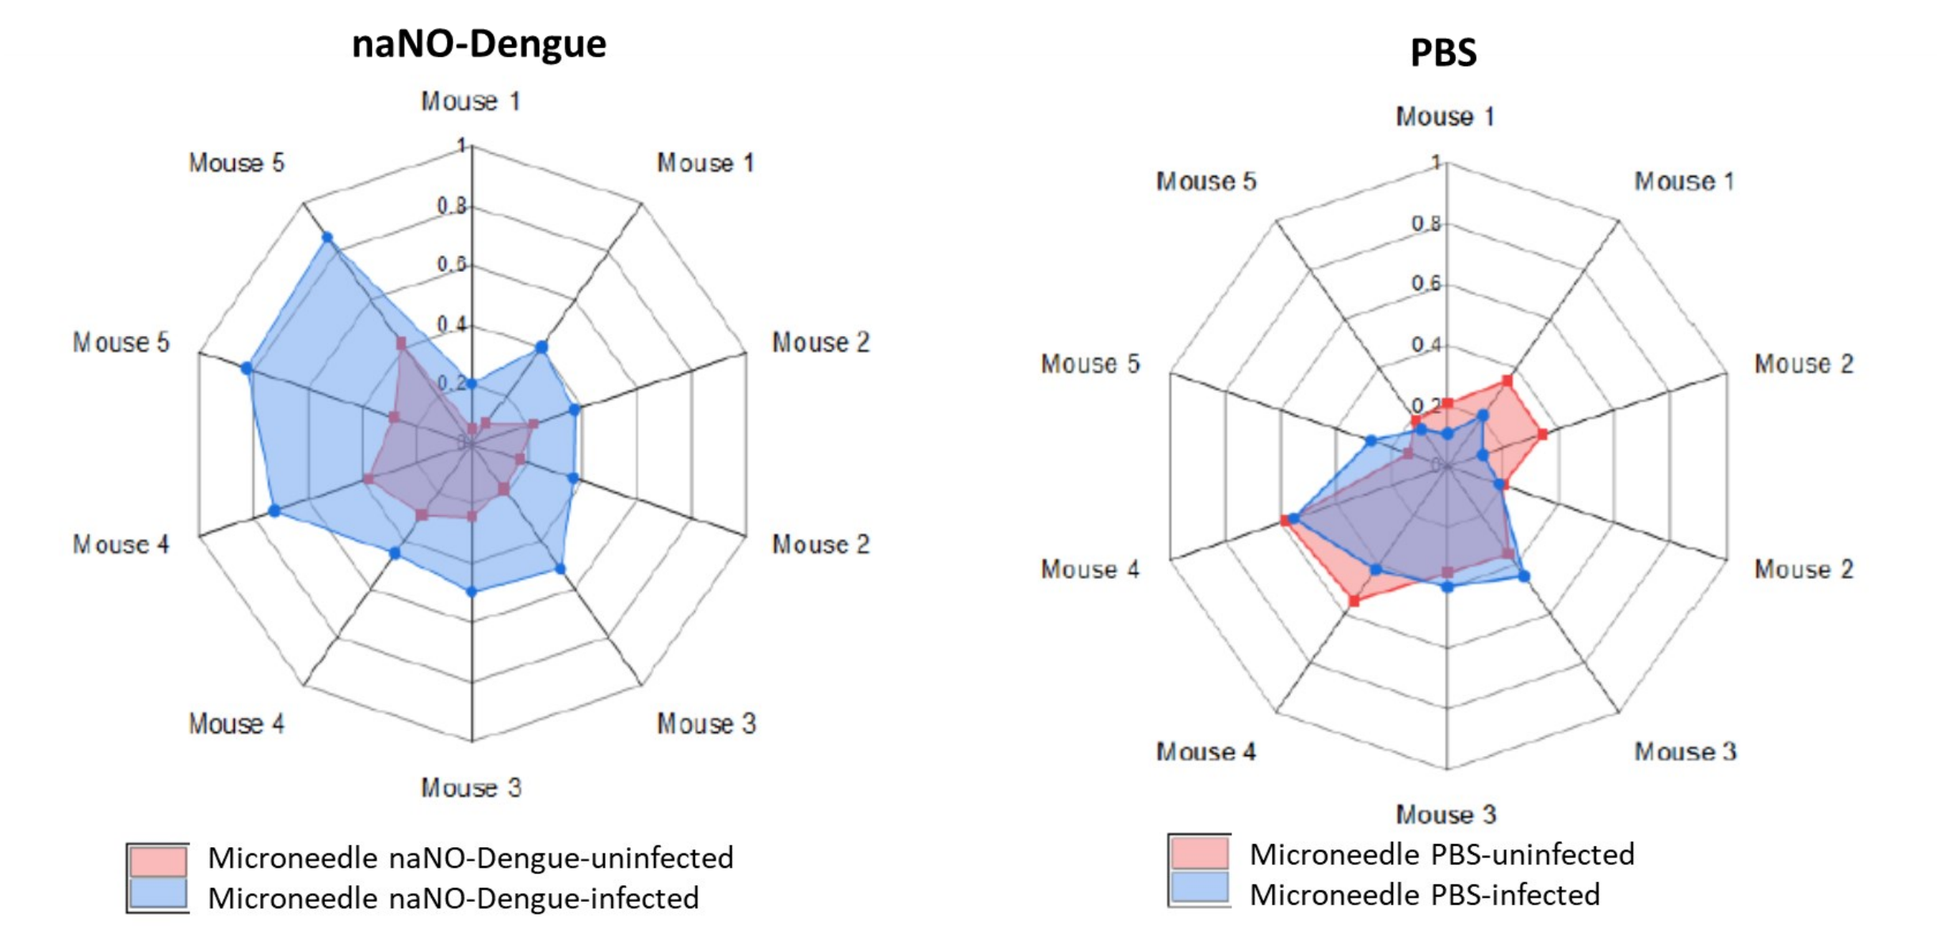
naNO-Dengue
PBS
Microneedle naNO-Dengue-uninfected
Microneedle naNO-Dengue-infected
Microneedle PBS-uninfected
Microneedle PBS-infected

Figure 6. Radar plots demonstrating the proportion of CD8+IFNγ+ cells in splenocytes isolated from mice vaccinated with the Dengue TCP vaccine formulation (left) or PBS (right) when co-cultured with either uninfected (pink) or Dengue-infected (blue) target cells at a T:E ratio of 1:1. Splenocytes were isolated and co-cultured with uninfected or Dengue-infected cells at a target (HepG2) to effector cell (T cells) ratio of 1:1 and the proportion of CD8+IFNγ+ cells was measured by flow cytometry.

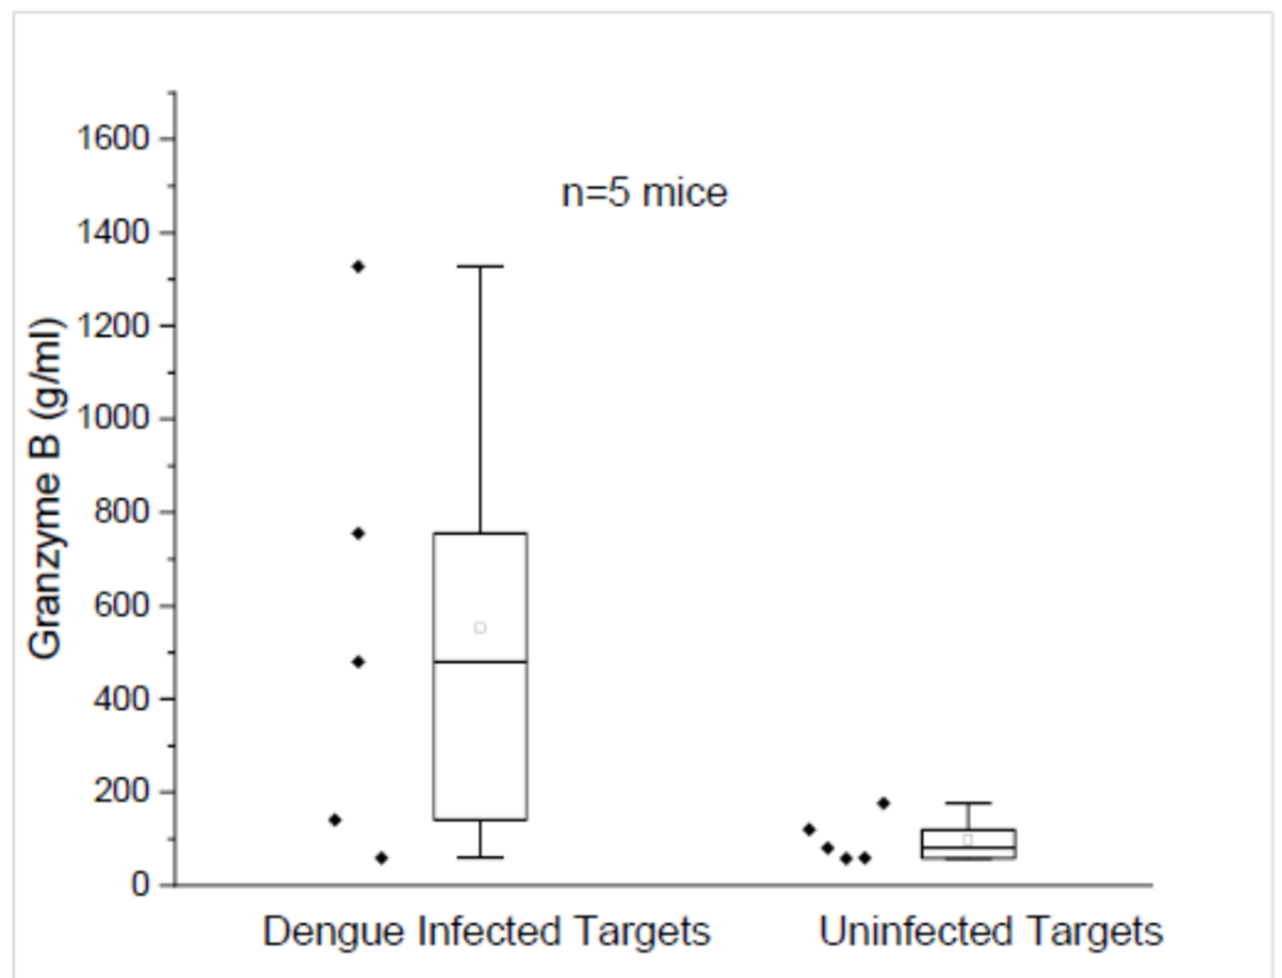
n=5 mice
Granzyme B (g/ml)
Dengue Infected Targets
Uninfected Targets

Figure 7. Granzyme B response of splenocytes isolated from transgenic HLA-A2 mice immunized with the Dengue TCP vaccine formulation via microneedle injection, then exposed to Dengue-infected or uninfected HepG2 cells at a T:E ratio of 0.1. Luminex magnetic bead cytokine assay (MAGPIX) was used to measure granzyme B level in the co-culture supernatant.

Conclusions

- Multiple **immunogenic**, virus **specific** candidate peptides were identified through mass spectrometry analysis of DENV infected cells.
- **Emergex's Dengue TCP vaccine** formulation primes a **T cell response** against DENV specific antigens ***in vivo*,** characterized by antigen-specific, functional **CD8+ T cells** that express CD107a, IFNɣ, and Granzyme B when evaluated *ex vivo*.
- A microneedle vaccine delivery method may support a robust immunological response increasing T cell effector function against viral infection.